The SOS1 Inhibitor MRTX0902 Demonstrates Activity Across Cancer Models with Mutations in Proximal Components of the RAS-MAPK Pathway

7268

Niranjan Sudhakar[1], **Larry Yan**[1], **Fadia Qiryaqos**[1], **Jade Laguer**[1], **David M Briere**[1], **Allan Hebbert**[1], **Andrew Calinisan**[1], **Silvia Coma**[2], **Jonathan Pachter**[2], **James G Christensen**[1], **Peter Olson**[1], **and Shilpi Khare**[1]

[1]Mirati, a Bristol Myers Squibb Company, San Diego, CA 92121, USA. [2]Verastem Oncology, Needham, MA 02494, USA.

BACKGROUND

- The RAS family of GTPases, which comprises KRAS, HRAS and NRAS, is mutated in ~19% of all human cancers and is associated with dysregulation of the RAS-MAPK pathway.[1]
- The Son of Sevenless proteins (SOS1 and SOS2) directly engage with the Switch I pocket of RAS and facilitate exchange of GDP to GTP, rendering the RAS protein active. As the prevalent SOS analog implicated in RAS activation, solely SOS1 is subject to adaptive feedback mechanisms within the RAS-MAPK pathway.[2,3]
- Functional genomics studies revealed that SOS1 depletion decreases the survival of KRAS-MAPK dependent cancer cells.[4] Consistently, SOS1 inhibitor treatment of preclinical human xenograft models with KRAS and MAPK pathway mutations resulted in tumor growth inhibition (TGI).[5,6]
- **MRTX0902** is a selective and orally bioavailable inhibitor of the SOS1:KRAS protein-protein interaction (PPI).[7] Administration of **MRTX0902** in combination with the KRAS G12C inhibitor, adagrasib, results in increased antitumor activity versus monotherapy in *KRAS* G12C mutant CDX and PDX models across various indications including NSCLC, PDAC, and CRC.[8] Furthermore, combined vertical inhibition of RAS-MAPK pathway signaling by **MRTX0902** with the EGFR inhibitor, osimertinib, results in improved antitumor activity in human tumor cells and CDX models harboring *EGFR* mutations.[8] **MRTX0902** is currently being evaluated in a Phase 1 clinical trials as a monotherapy and combination partner of adagrasib in patients with advanced solid tumors with mutations in the KRAS-MAPK pathway.
- While there are approved targeted therapies for patients with mutations in *KRAS* G12C in metastatic NSCLC, **there remains a need for effective therapy for cancers driven by RAS-MAPK pathway mutations:**
 - GTPase-activating proteins (GAPs) negatively regulate RAS activation by promoting conversion of GTP-RAS to GDP-RAS. Genetic alterations in the GAP, neurofibromin (***NF1***), are prevalent in nearly 7% of all cancers with greatest incidence in lung and colon adenocarcinoma and at a lower frequency in the inherited neurofibromatosis type 1 disorder.[9,10]
 - Activating ***SOS1*** mutations are found in approximately 1% of NSCLC cases and up to 10% in RASopathies including Noonan's Syndrome and gingival fibromatosis.[11,12,13]
 - Gain of function mutations in the Src homology-2 domain-containing phosphatase 2 ***(SHP2)*** adaptor protein, encoded by the *PTPN11* gene, have been implicated in Juvenile leukemia, neuroblastoma, and developmental disorders including LEOPARD Syndrome.[14,15]
 - In contrast to the class I and class II *BRAF* mutations, class III mutations in *BRAF* are dependent on GTP loaded RAS to drive ERK1/2 signalling.[14]
- The RAF/MEK clamp, avutometinib (Verastem Oncology), blocks both RAF and MEK kinase activities, thereby preventing reactivation of MEK by RAF and leading to antitumor activity in RAS/MAPK-dependent cancers. Inhibition of MEK leads to a release of a negative feedback loop, resulting in increased SOS1 activity. We hypothesized that inhibition of RAF, MEK and SOS1 could lead to greater and durable antiproliferative activity in tumors with MAPK activation.
- In the present studies, we have shown that **MRTX0902** demonstrates single agent activity in human tumor cells and xenograft models with mutations in *NF1, PTPN11, SOS1* and class III *BRAF* by modulating ERK1/2 phosphorylation. Additionally, combination with the RAF/MEK clamp avutometinib demonstrated greater antitumor activity versus monotherapy treatment.

Figure 1. MRTX0902 combination with avutometinib results in durable RAS-MAPK pathway suppression

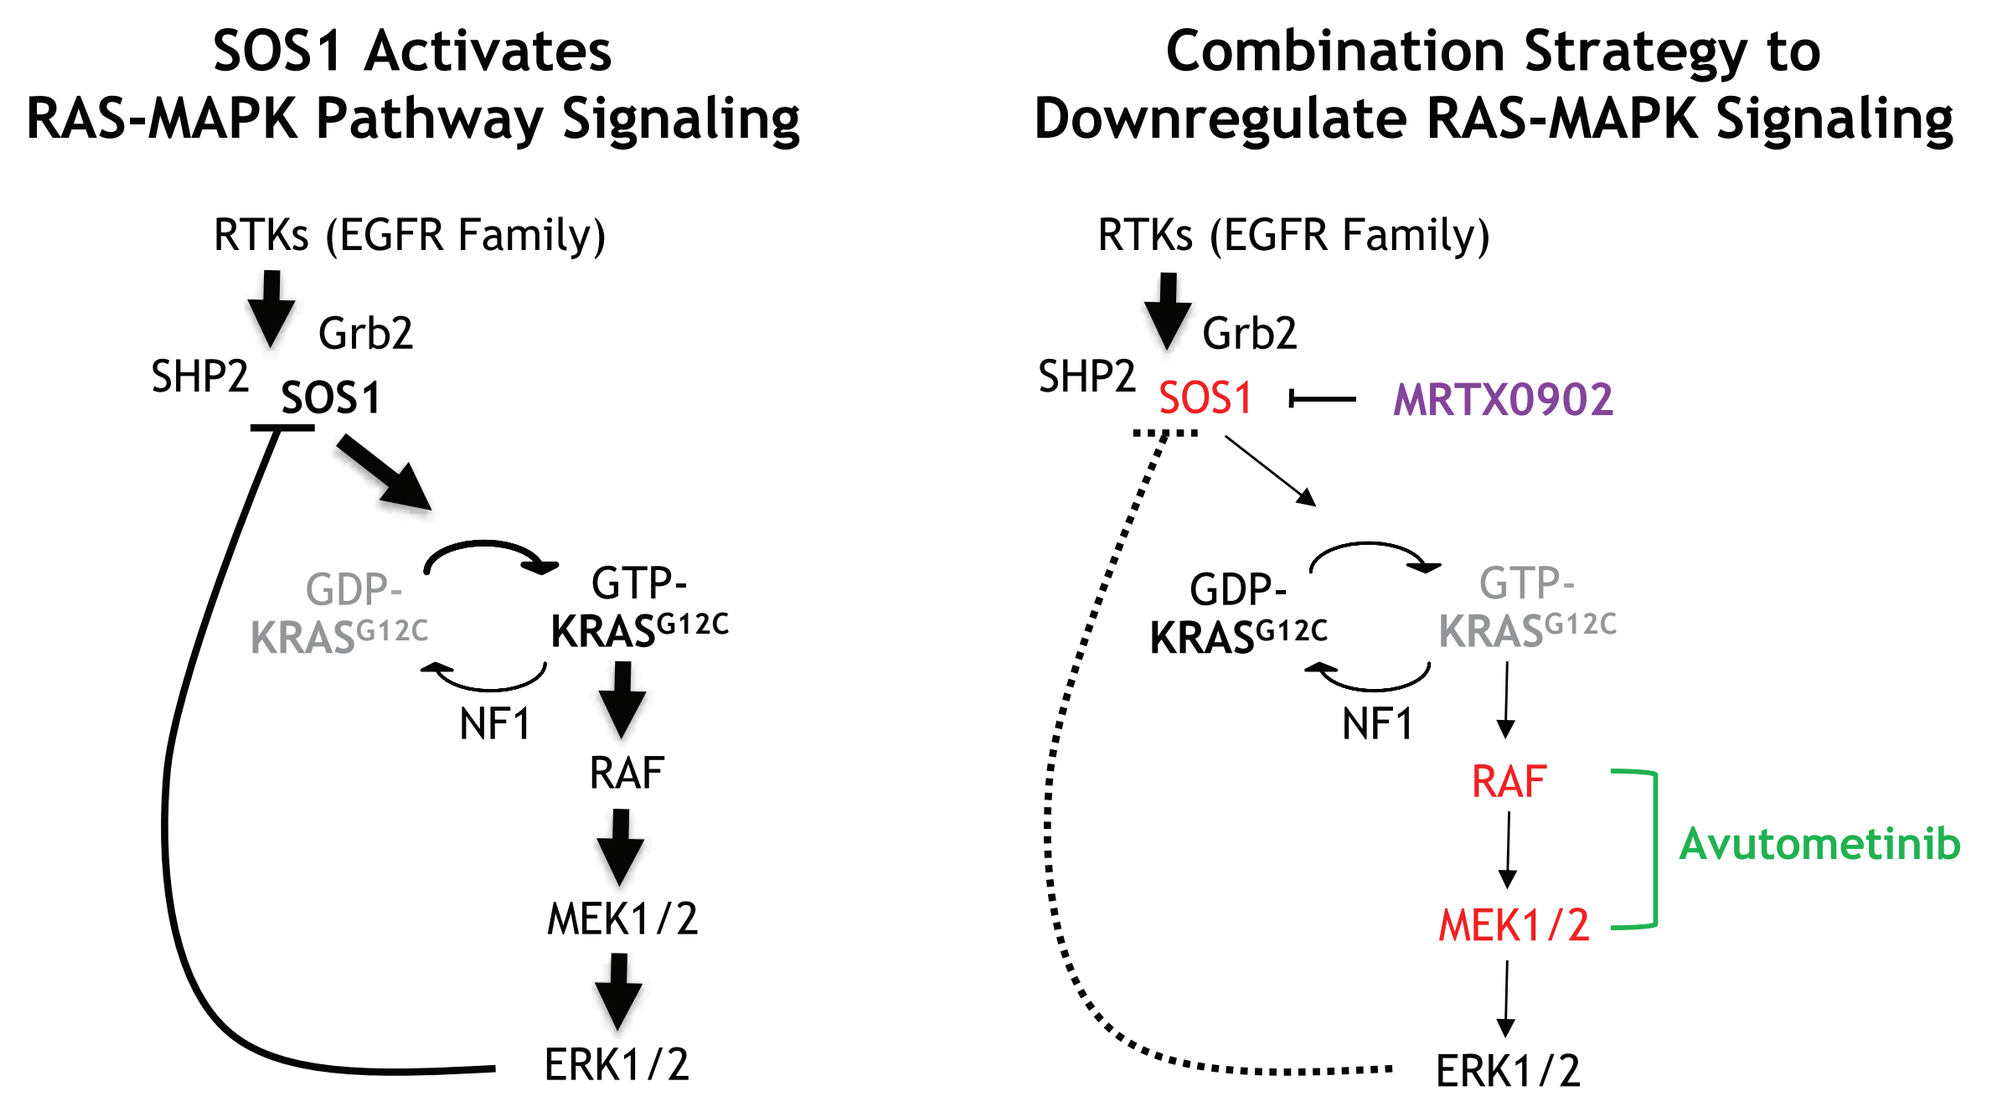

SOS1 Activates
RAS-MAPK Pathway Signaling
RTKs (EGFR Family)
Grb2
SHP2
SOS1
GDP-KRAS^{G12C}
GTP-KRAS^{G12C}
NF1
RAF
MEK1/2
ERK1/2
Combination Strategy to
Downregulate RAS-MAPK Signaling
RTKs (EGFR Family)
Grb2
SHP2
SOS1
MRTX0902
GDP-KRAS^{G12C}
GTP-KRAS^{G12C}
NF1
RAF
MEK1/2
Avutometinib
ERK1/2

METHODS

- ***In vitro* pERK assay:** The cellular activity of **MRTX0902** (inhibition of ERK1/2 phosphorylation) was evaluated in the In-Cell Western (ICW) assay. Cells were incubated with serial dilutions of MRTX0902 for 30 minutes prior to probing with primary antibodies for pERK1/2 and GapDH and IRDye® secondary antibodies.
- ***In vitro* cell viability assay:** Cells were seeded in a special ultra-low attachment (ULA) microplate to allow for 3D spheroid formation. After 24 hours, cells were dosed with either serial dilutions of MRTX0902 as a single agent or in combination with avutometinib for synergy experiments. Following 7-14-days, CellTiter-Glo® (CTG) reagent was added, and the luminescence was read using the CLARIOstar® microplate reader.
- ***In vivo* Mice Studies:**
 - Tumor growth inhibition (TGI) studies: Human tumor cells were subcutaneously injected into the right flank of mice to establish human tumor cell-line derived (CDX) models. When the average tumor volumes reached the desired starting volume, animals were dosed with vehicle, MRTX0902, and/or avutometinib as described. Tumor size and animal body weights were measured twice per week. Tumor growth inhibition between two experimental groups was determined to be statistically significant using the two-tailed Student's *t* test. Brackets indicate *p*-value <0.05 (*), *p*-value <0.005 (**), *p*-value <0.0005 (***), or *p*-value <0.0001 (****).
- **Pharmacodynamic (PD) studies:** Tumors were collected 7 days post dosing of either vehicle, MRTX0902, and/or avutometinib in human tumor cell-line derived (CDX) models and processed for Western Blot analysis and RAS pathway protein levels were assessed as described. Reduction of pERK or p4EBP1 relative fluorescence intensity between treatment groups was determined to be statistically significant using the two-tailed Student's *t* test. Brackets indicate *p*-value <0.05 (*), *p*-value <0.005 (**), *p*-value <0.0005 (***), or *p*-value <0.0001 (****).

RESULTS

MRTX0902 modulates RAS-dependent signaling and inhibits growth of human tumor models harboring mutations in the RAS-MAPK pathway

- Significant inhibition of pERK and 3D cell viability was observed in *EGFR* mutant (NCI-H1975 and PC9), *PTPN11* mutant (LN229), *SOS1* mutant (OCI-AML5), *NF1* mutant (HCC1438) and the class III *BRAF* (NCI-H508 and NCI-H1666) cell lines with IC_{50} values <250 nM.
- MRTX0902 treatment resulted in significant tumor growth inhibition and decreases in pERK levels in RL95-2 (*SOS1* N233I) and MKN74 (*NF1* X547) CDX models.
- MRTX0902 treatment led to notable *in vivo* activity in human tumor cell line-derived (CDX) models harboring mutations in the upstream members of the MAPK pathway.

Figure 2. MRTX0902 demonstrates antiproliferative activity in a panel of RAS-MAPK pathway-mutant human tumor cell lines

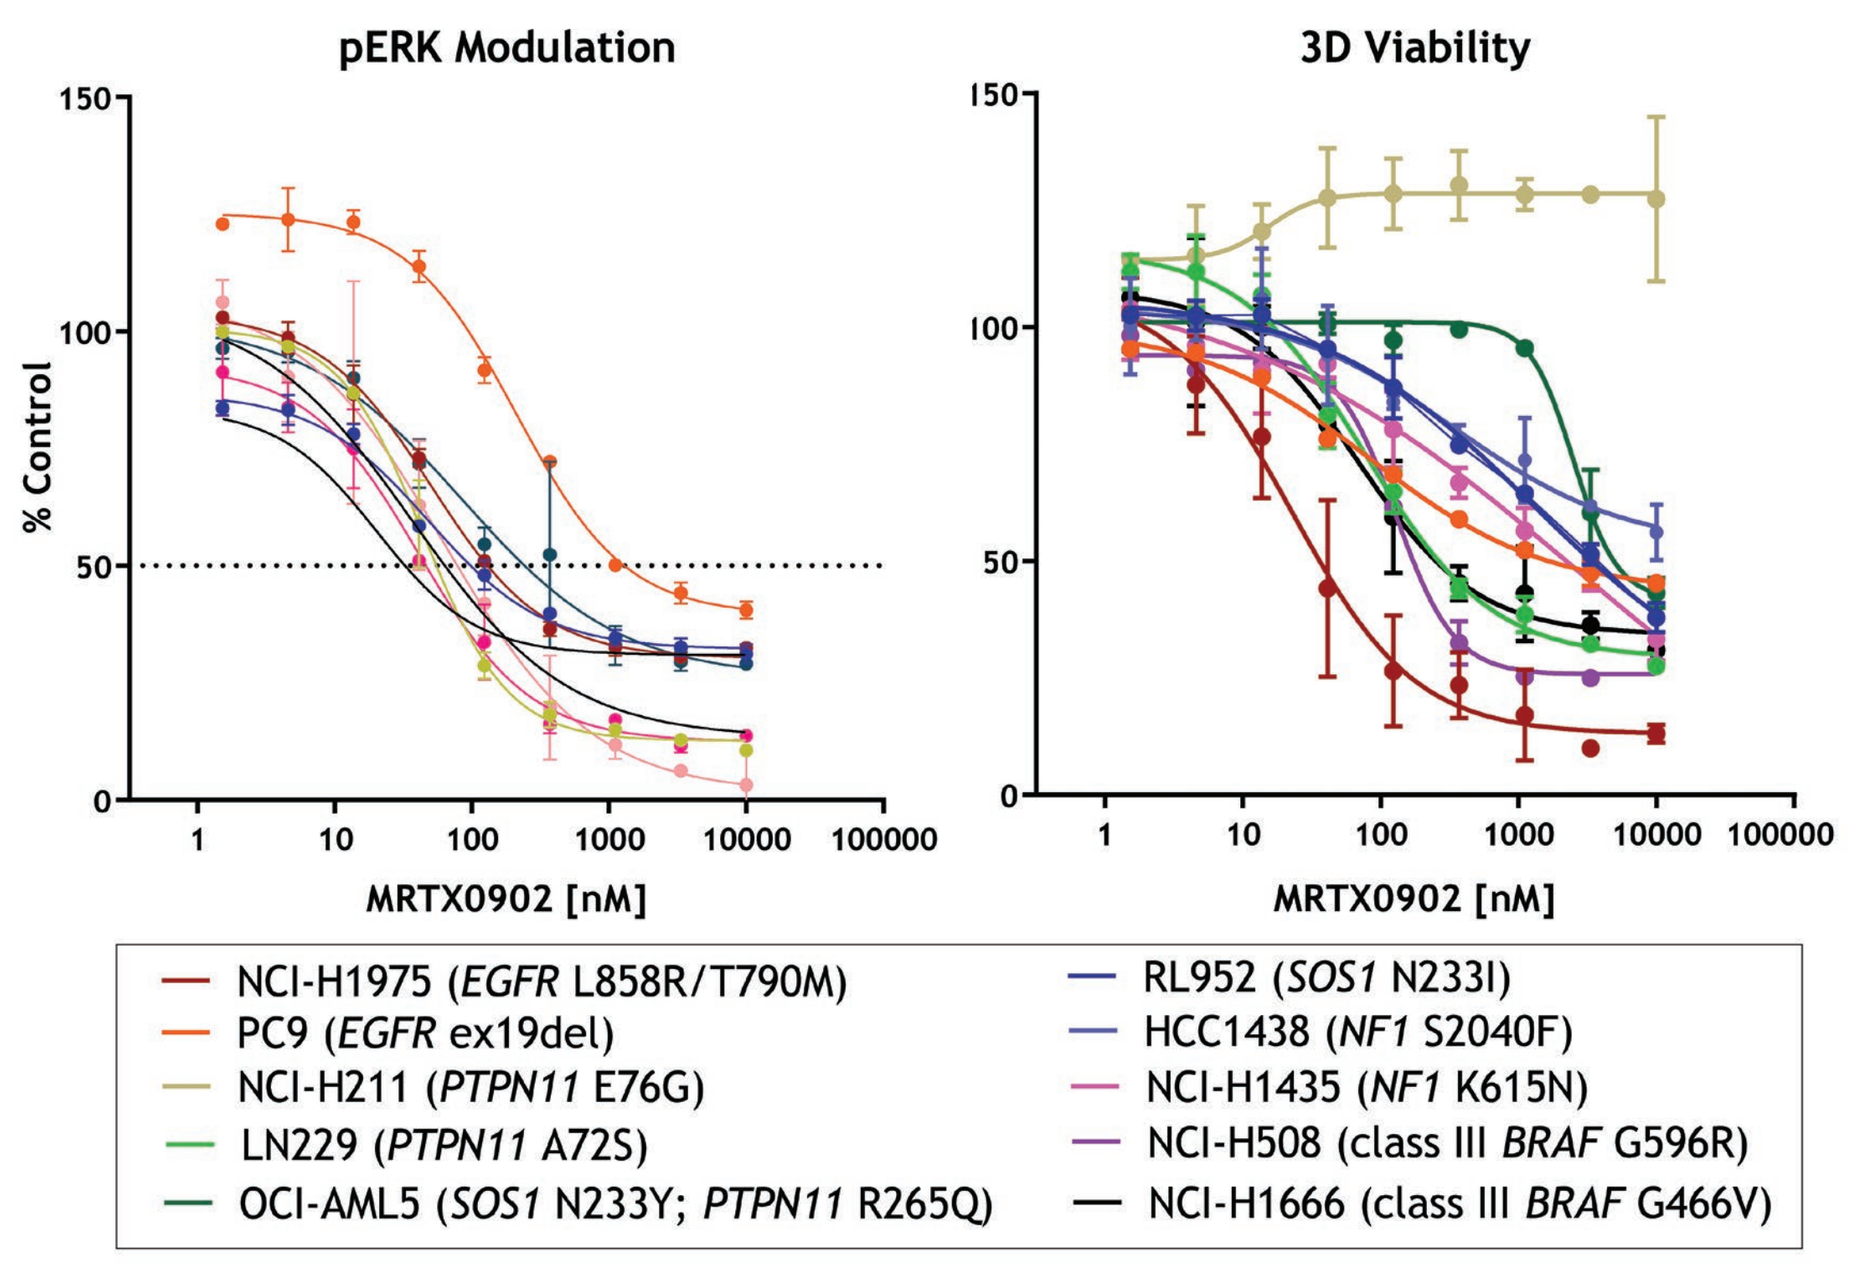

pERK Modulation
3D Viability
% Control
MRTX0902 [nM]
NCI-H1975 (EGFR L858R/T790M)
PC9 (EGFR ex19del)
NCI-H211 (PTPN11 E76G)
LN229 (PTPN11 A72S)
OCI-AML5 (SOS1 N233Y; PTPN11 R265Q)
RL952 (SOS1 N233I)
HCC1438 (NF1 S2040F)
NCI-H1435 (NF1 K615N)
NCI-H508 (class III BRAF G596R)
NCI-H1666 (class III BRAF G466V)

Figure 3. MRTX0902 monotherapy treatment leads to antitumor activity in *SOS1*-mutant and *NF1*-mutant CDX models

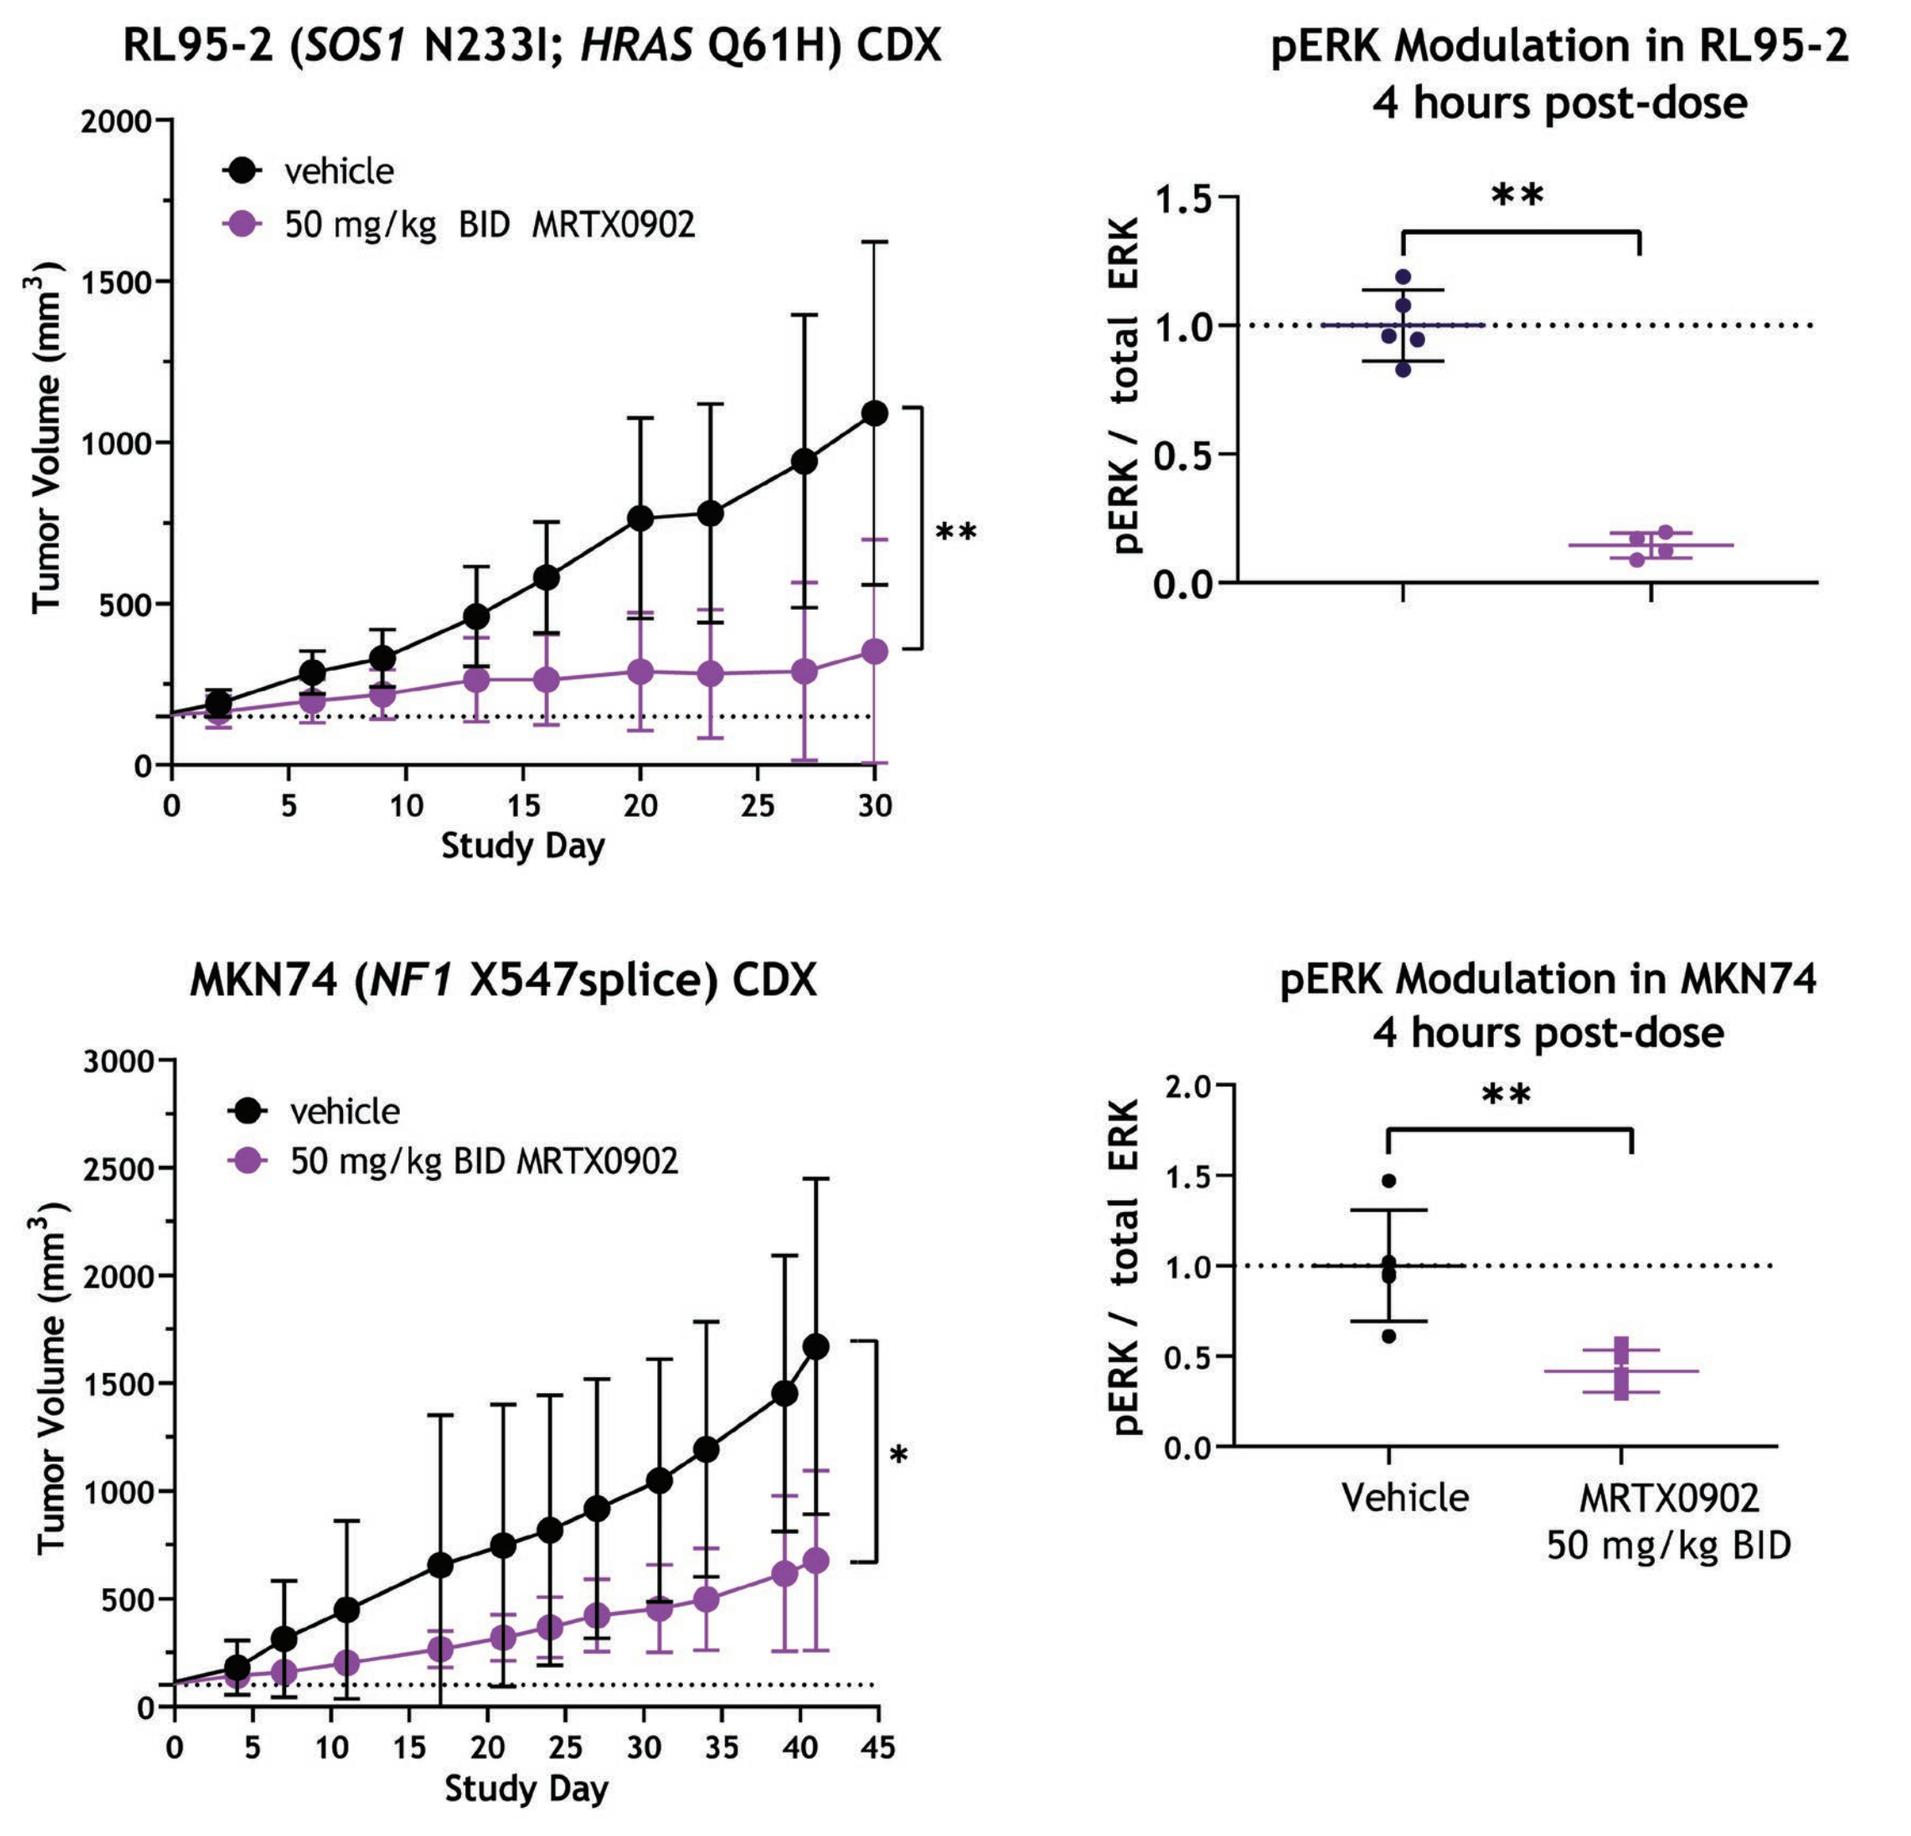

RL95-2 (SOS1 N233I; HRAS Q61H) CDX
vehicle
50 mg/kg BID MRTX0902
Tumor Volume (mm3)
Study Day
pERK Modulation in RL95-2
4 hours post-dose
pERK / total ERK
MKN74 (NF1 X547splice) CDX
pERK Modulation in MKN74
4 hours post-dose
Vehicle
MRTX0902
50 mg/kg BID

Figure 4. ***In vivo*** **breadth of efficacy for MRTX0902 in RAS-MAPK pathway-mutant human tumor xenograft models**

MRTX0902 was administered twice daily via oral gavage for ~14-68 days to immunocompromised mice bearing human RAS-MAPK pathway-mutant tumor xenografts (average starting tumor volume of 100-200 mm³) at 50 mg/kg.

MRTX0902 was formulated in a suspension of 0.5% methylcellulose (4000 cps) + 0.2% Tween 80 in water. Data for n=4 or 5 animals/group is shown as percent change in baseline tumor volume.

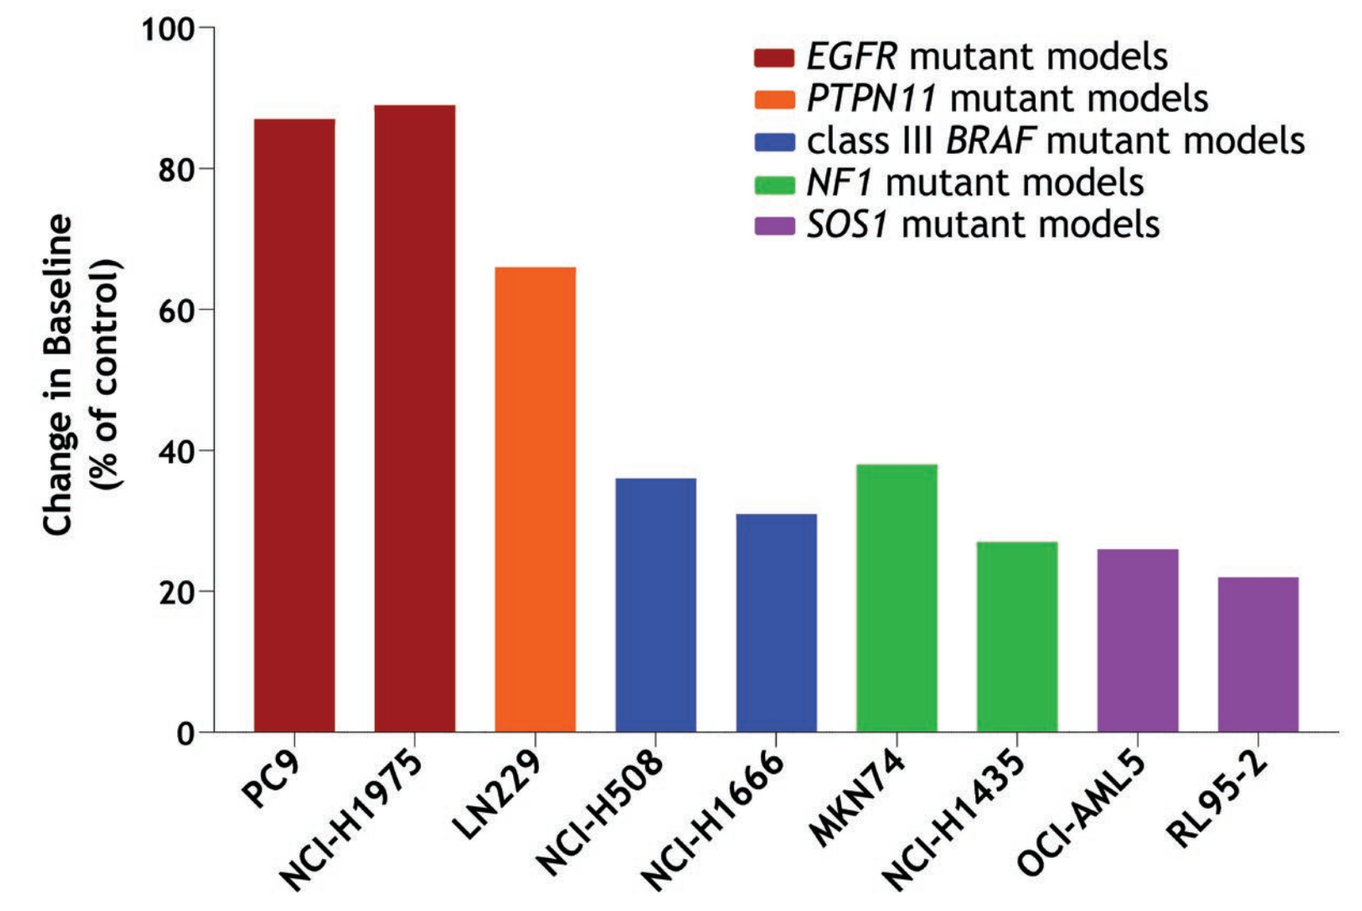

Change in Baseline (% of control)
EGFR mutant models
PTPN11 mutant models
class III BRAF mutant models
NF1 mutant models
SOS1 mutant models
PC9
NCI-H1975
LN229
NCI-H508
NCI-H1666
MKN74
NCI-H1435
OCI-AML5
RL95-2

MRTX0902 with avutometinib exhibits greater anti-proliferative activity and suppresses RAS-MAPK signaling

Figure 5. The combination of MRTX0902 with avutometinib leads to more durable downregulation of RAS-MAPK pathway signaling versus avutometinib monotherapy *in vitro*

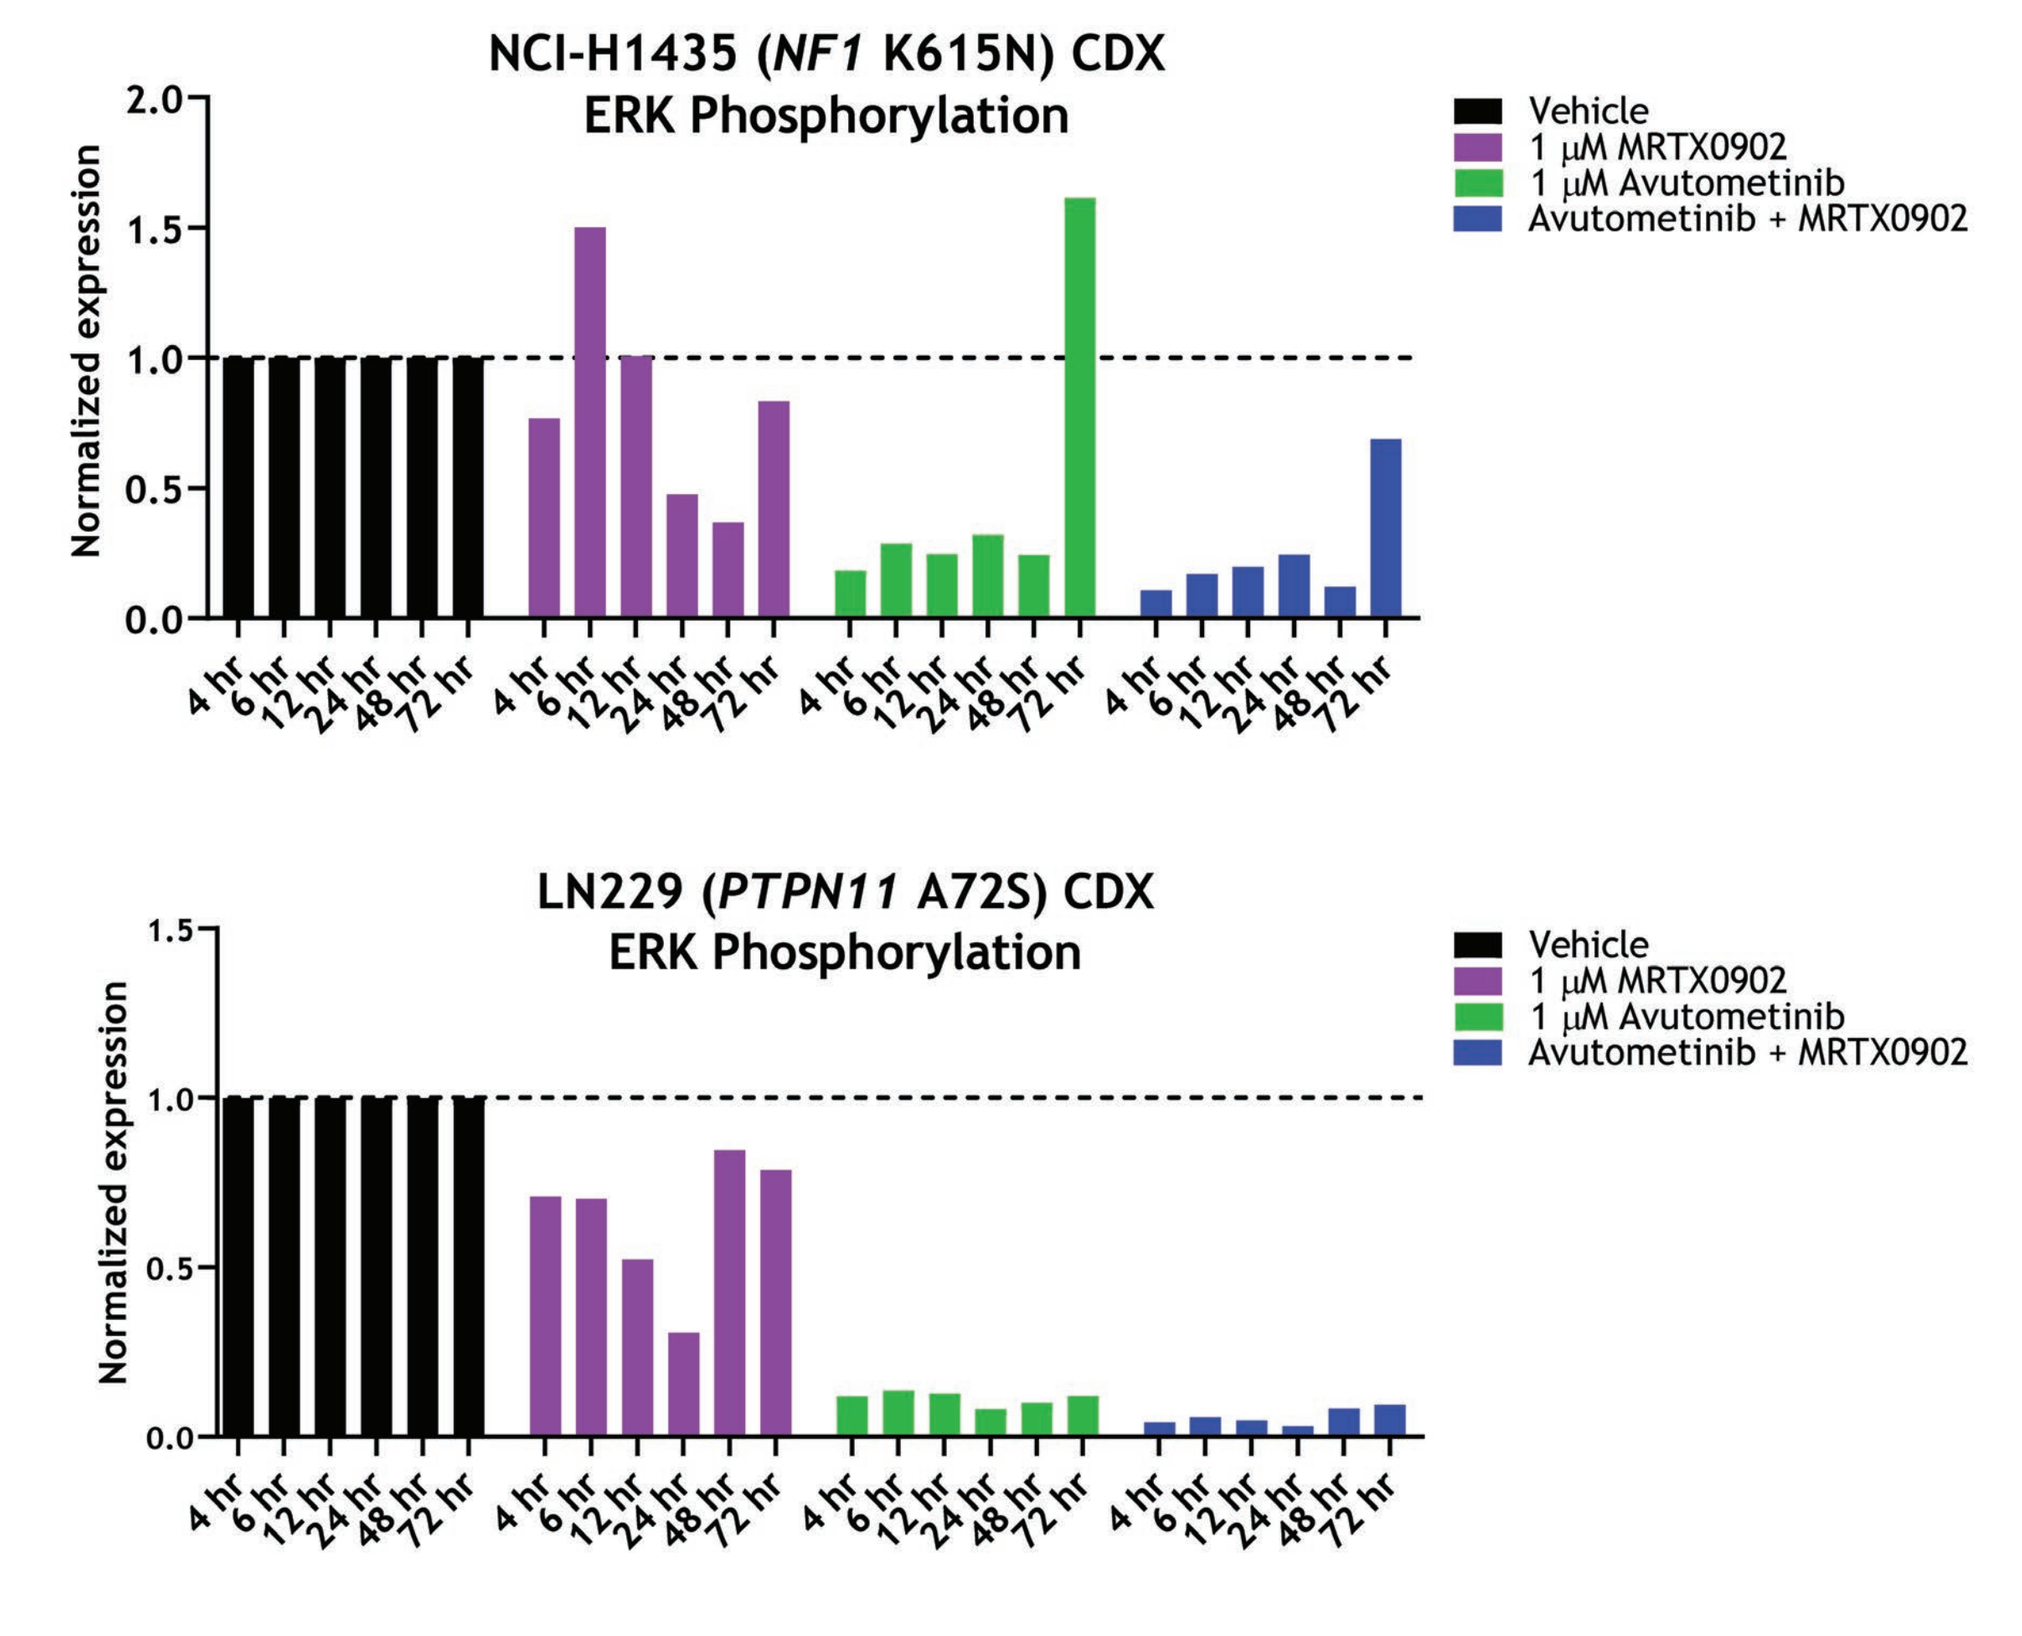

NCI-H1435 (NF1 K615N) CDX
ERK Phosphorylation
LN229 (PTPN11 A72S) CDX
ERK Phosphorylation
Normalized expression
Vehicle
1 µM MRTX0902
1 µM Avutometinib
Avutometinib + MRTX0902

- MRTX0902 combination with avutometinib demonstrated greater pERK inhibition versus avutometinib or MRTX0902 monotherapy in the LN229 (*PTPN11* A72S) and NCI-H1435 (*NF1* K615N) cell lines.

Figure 6. Combination of MRTX0902 and avutometinib leads to synergistic effects *in vitro*

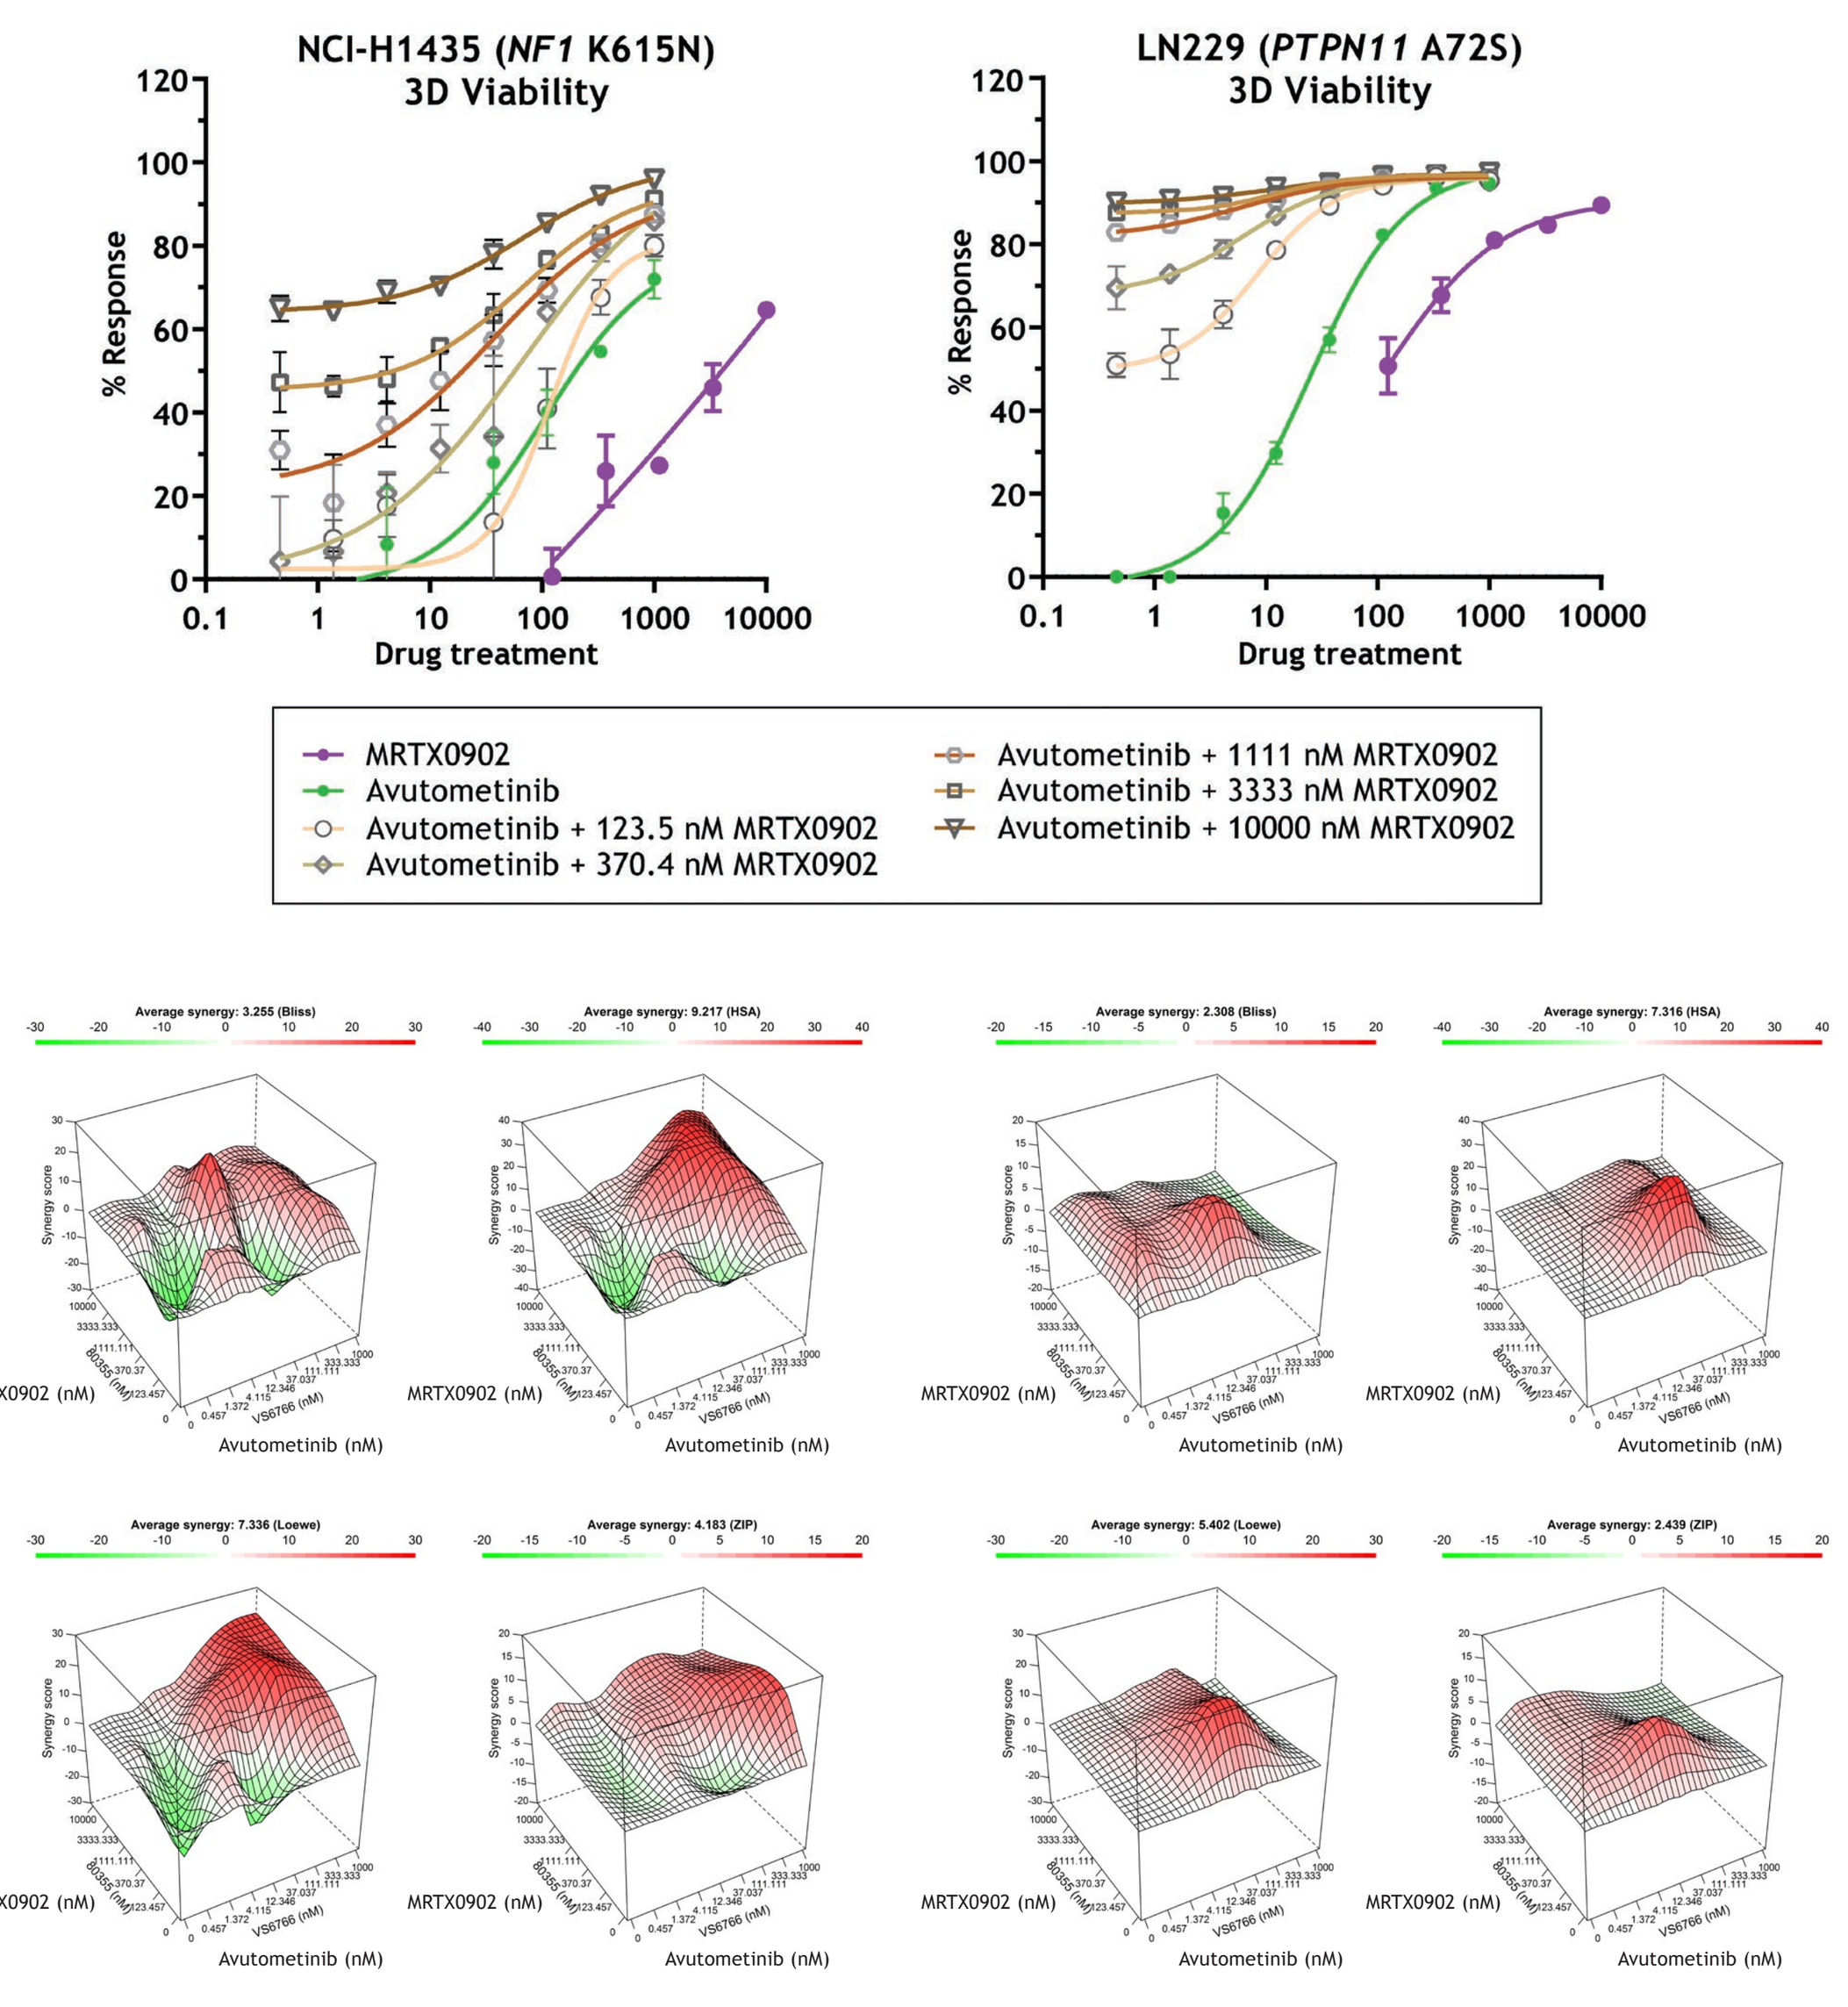

NCI-H1435 (NF1 K615N)
3D Viability
LN229 (PTPN11 A72S)
3D Viability
% Response
Drug treatment
MRTX0902
Avutometinib
Avutometinib + 123.5 nM MRTX0902
Avutometinib + 370.4 nM MRTX0902
Avutometinib + 1111 nM MRTX0902
Avutometinib + 3333 nM MRTX0902
Avutometinib + 10000 nM MRTX0902

Table 1. Inhibitory effects on NCI-H1435 (*NF1* K615N) and LN229 (*PTPN11* A72S) 3D cell proliferation following 14 days of drug treatment was assessed via the CellTiter-Glo® assay. A custom R-script was used to generate a composite synergy score (Mirati Combination Analysis or MCA) for combination treatment of both drugs tested at a 5-point or 8-point dose-response curve.[16]

		Synergy Scores with MRTX0902				
Cell line	**Combination Partner**	**Bliss**	**HSA**	**Loewe**	**Zip**	**MCA**
NCI-H1435	Avutometinib	3.3	9.2	7.3	4.2	24
LN229	Avutometinib	2.3	7.3	5.4	2.4	17.4

MRTX0902 with avutometinib demonstrates a more durable antitumor effect

Figure 7. MRTX0902 with avutometinib leads to significantly greater suppression of RAS-MAPK pathway signaling versus avutometinib or MRTX0902 monotherapy *in vivo*

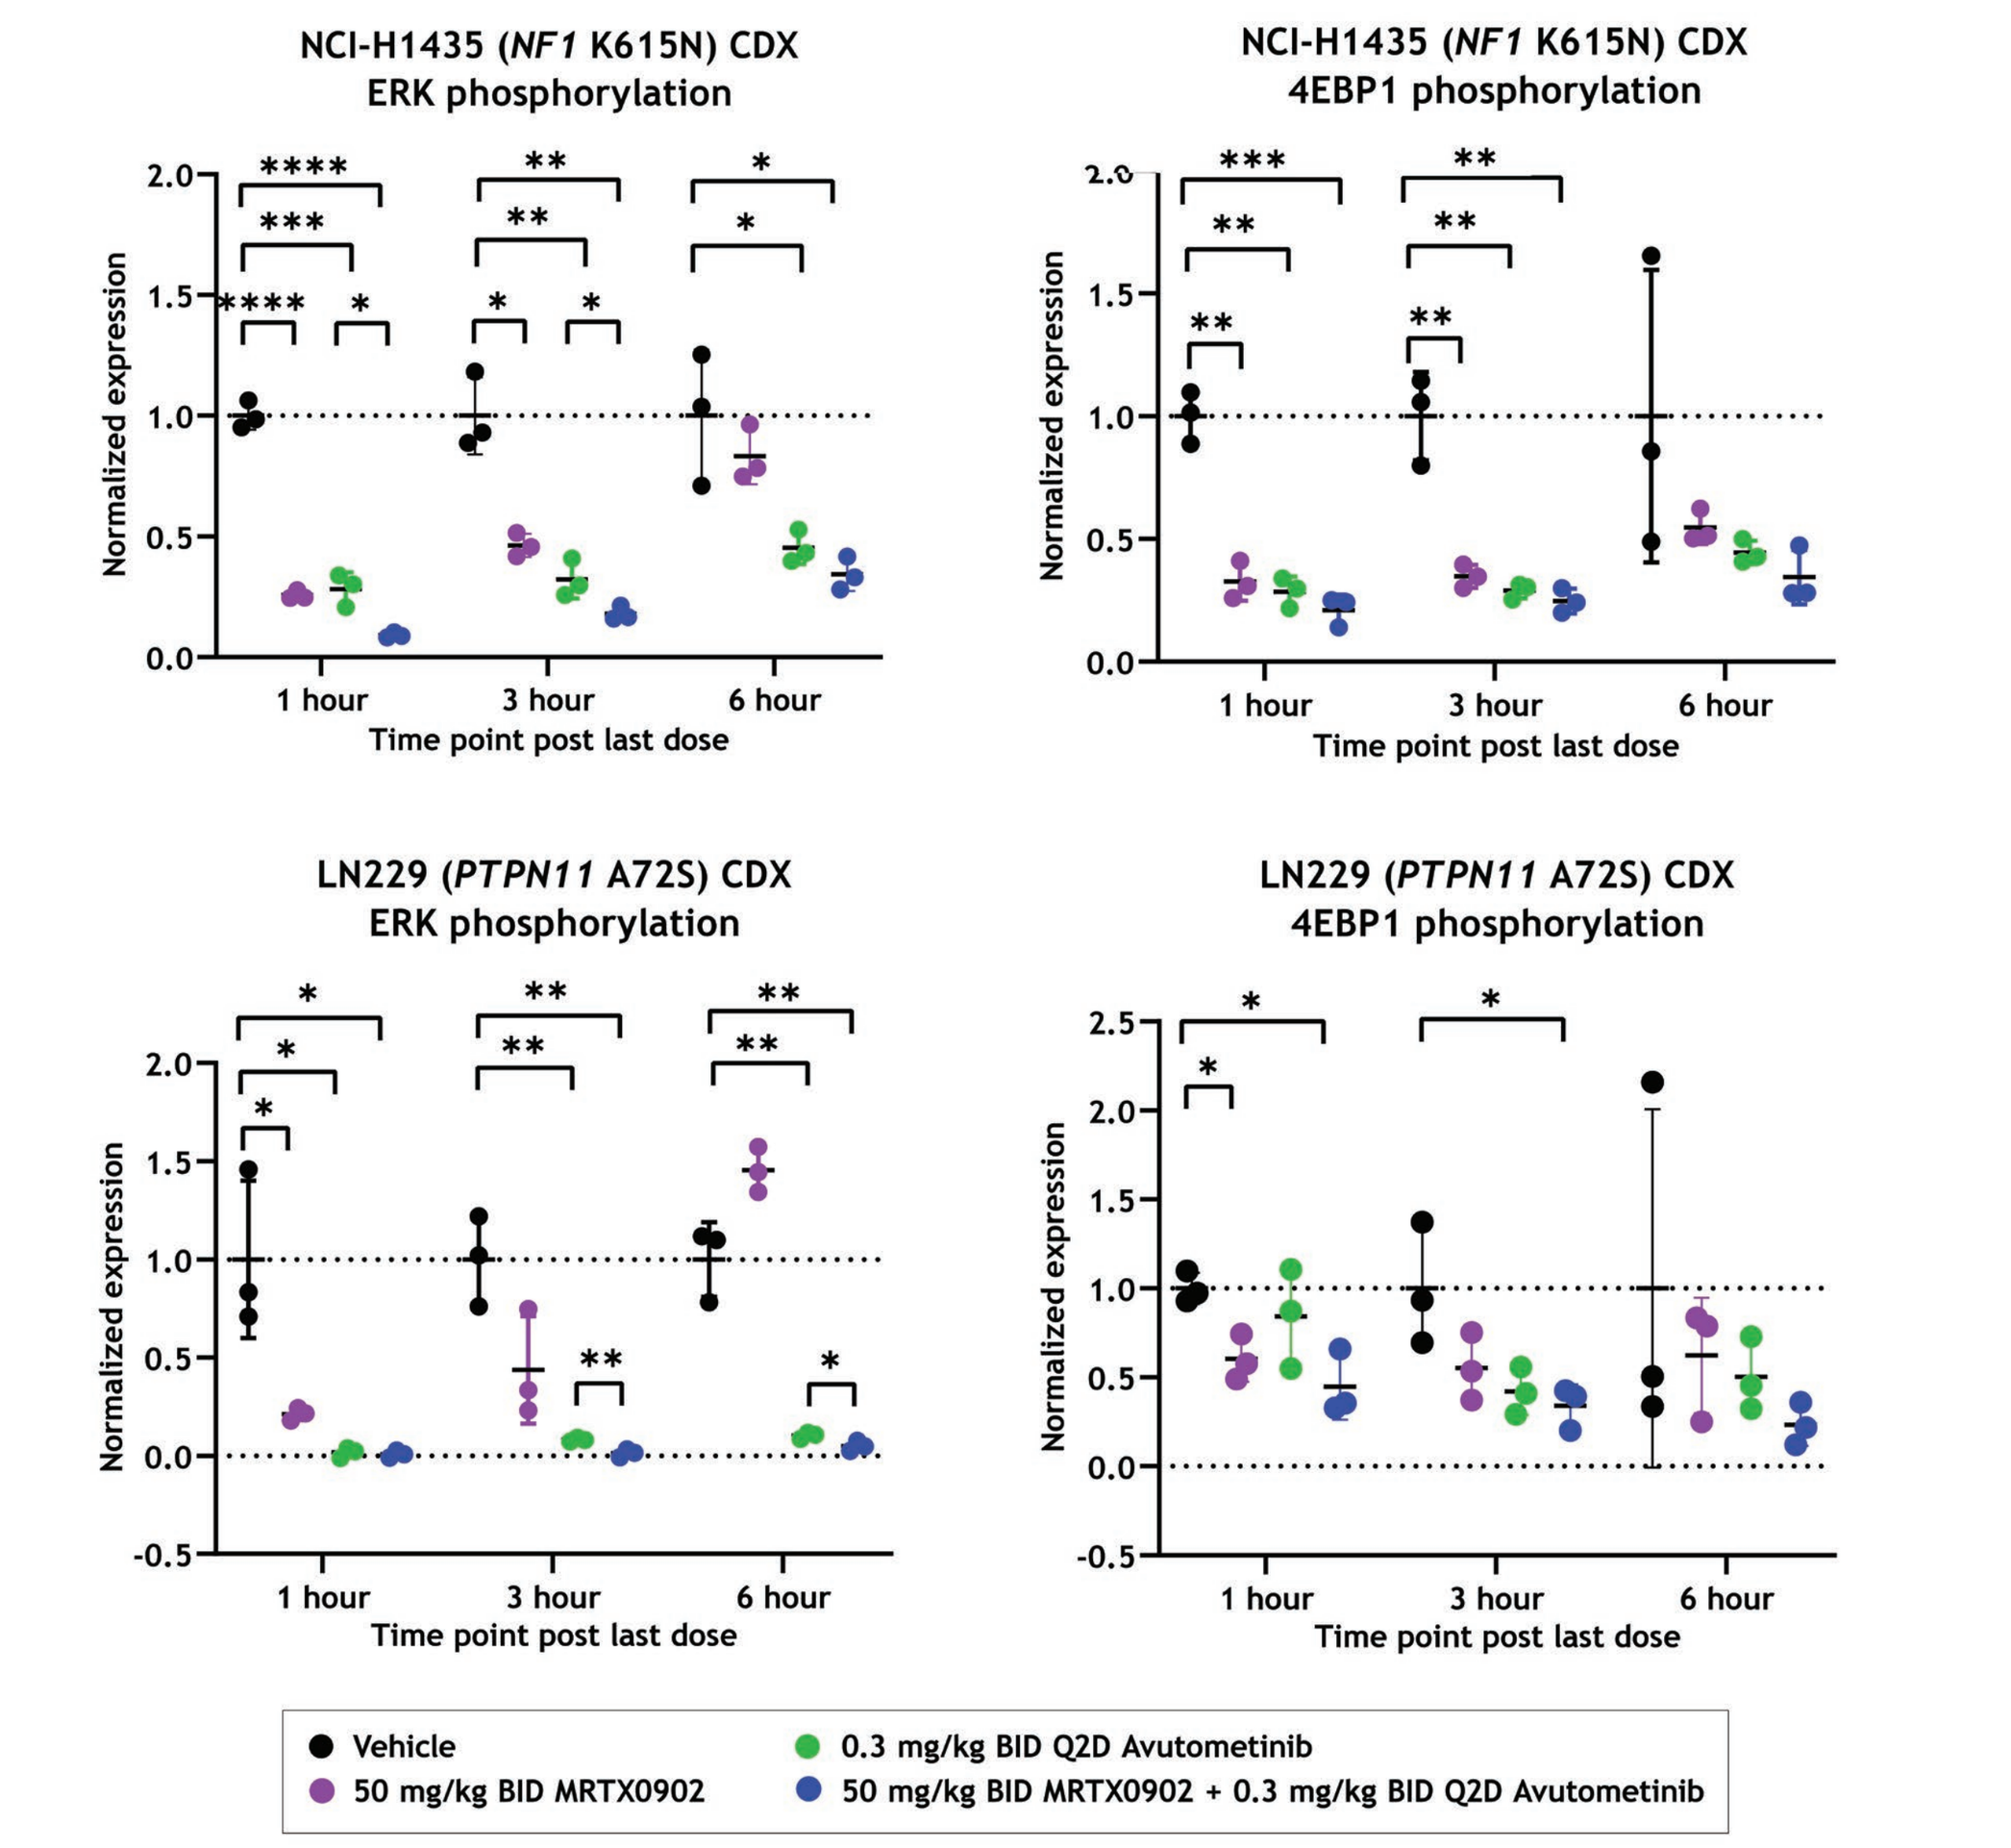

NCI-H1435 (NF1 K615N) CDX
ERK phosphorylation
NCI-H1435 (NF1 K615N) CDX
4EBP1 phosphorylation
LN229 (PTPN11 A72S) CDX
ERK phosphorylation
LN229 (PTPN11 A72S) CDX
4EBP1 phosphorylation
Normalized expression
Time point post last dose
Vehicle
50 mg/kg BID MRTX0902
0.3 mg/kg BID Q2D Avutometinib
50 mg/kg BID MRTX0902 + 0.3 mg/kg BID Q2D Avutometinib

- Combination treatment of MRTX0902 with avutometinib led to significant reductions in pERK levels versus avutometinib or MRTX0902 monotherapy treatment at 1- and 3-hours post treatment in the NCI-H1435 (*NF1* K615N) model and at 3- and 6-hours post treatment in the LN229 (*PTPN11* A72S) model.

Figure 8. Co-administration of MRTX0902 with avutometinib leads to greater depth of antitumor activity versus avutometinib monotherapy in *NF1*-, *PTPN11*-, and class III *BRAF*-mutant xenograft models

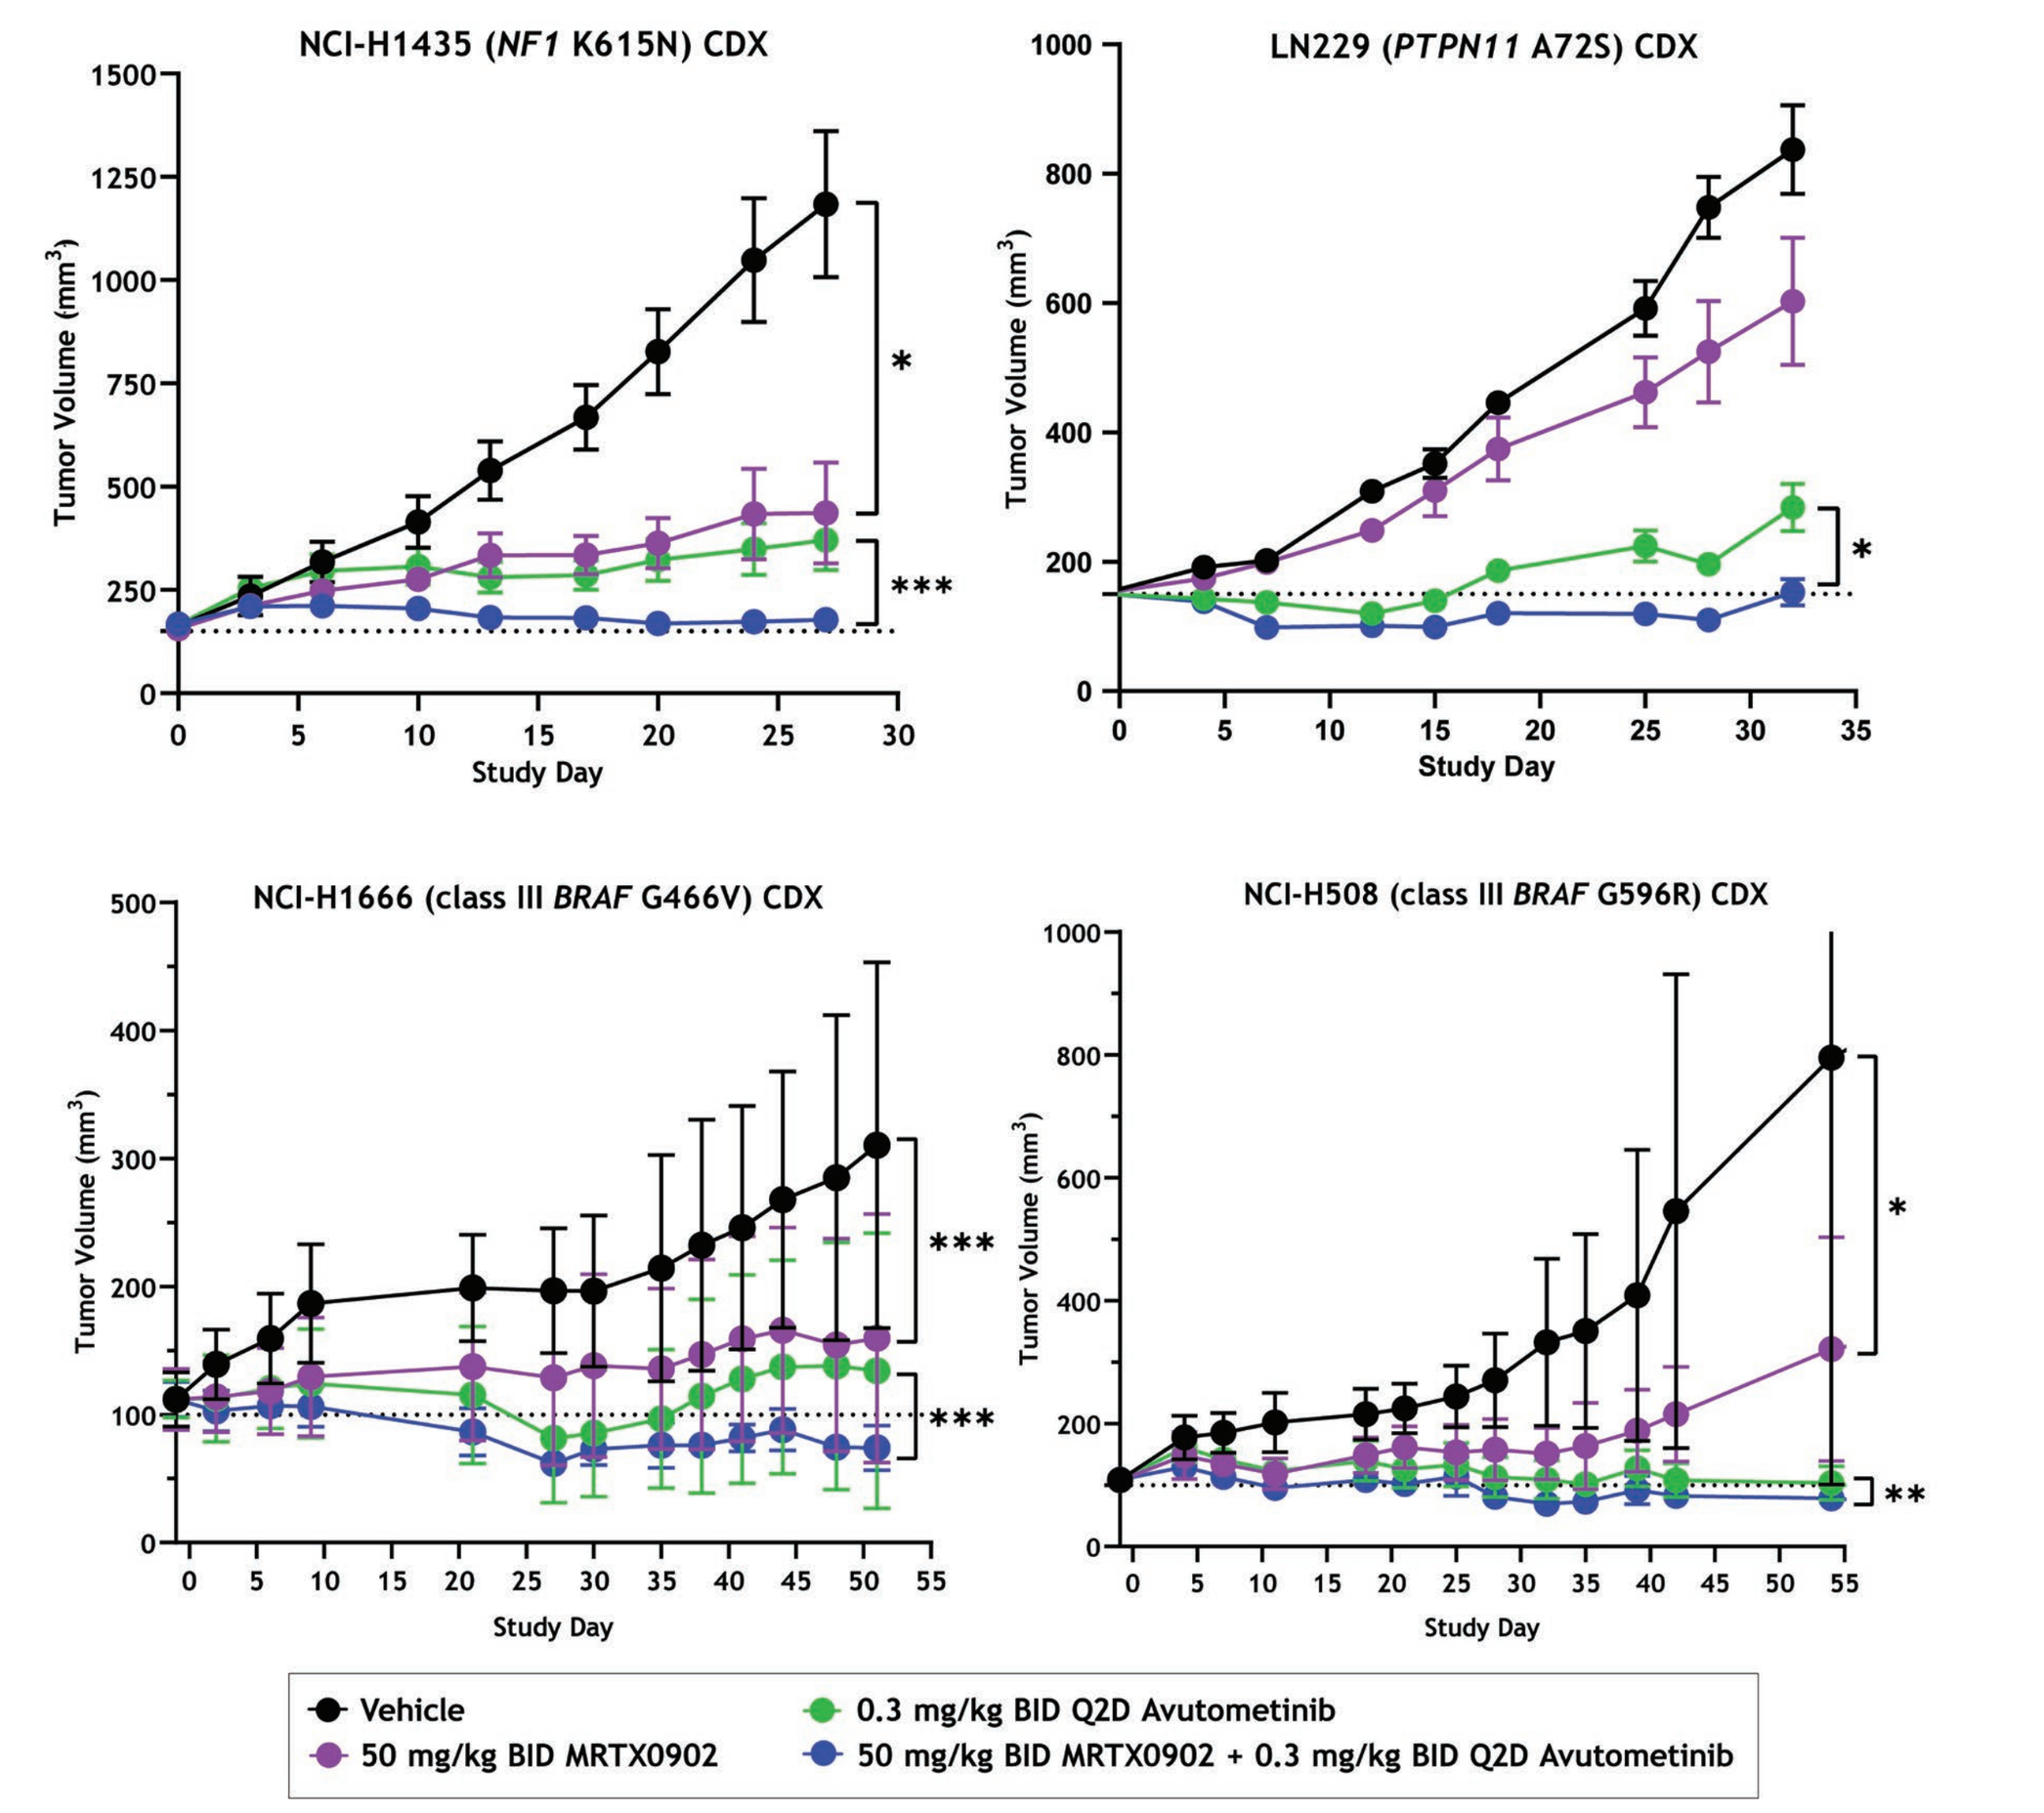

NCI-H1435 (NF1 K615N) CDX
LN229 (PTPN11 A72S) CDX
NCI-H1666 (class III BRAF G466V) CDX
NCI-H508 (class III BRAF G596R) CDX
Tumor Volume (mm3)
Study Day
Vehicle
50 mg/kg BID MRTX0902
0.3 mg/kg BID Q2D Avutometinib
50 mg/kg BID MRTX0902 + 0.3 mg/kg BID Q2D Avutometinib

Table 2: MRTX0902 enhances the antitumor effect of avutometinib in CDX models

Model	Tumor Type	MRTX0902 Treatment	Avutometinib Treatment	% Growth Inhibition/ Regression (Day)	Response
NCI-H1435	NSCLC	50 mg/kg BID	n/a	73% (27)	TGI
		n/a	0.3 mg/kg BID Q2D	80% (27)	TGI
		50 mg/kg BID	0.3 mg/kg BID Q2D	99% (27)	TGI
NCI-H1666	NSCLC	50 mg/kg BID	n/a	69% (51)	TGI
		n/a	0.3 mg/kg BID Q2D	89% (51)	TGI
		50 mg/kg BID	0.3 mg/kg BID Q2D	-34% (51)	Regression

Figure 9. ***In vivo*** **breadth of efficacy for MRTX0902 and avutometinib in RAS-MAPK pathway-mutant human tumor xenograft models**

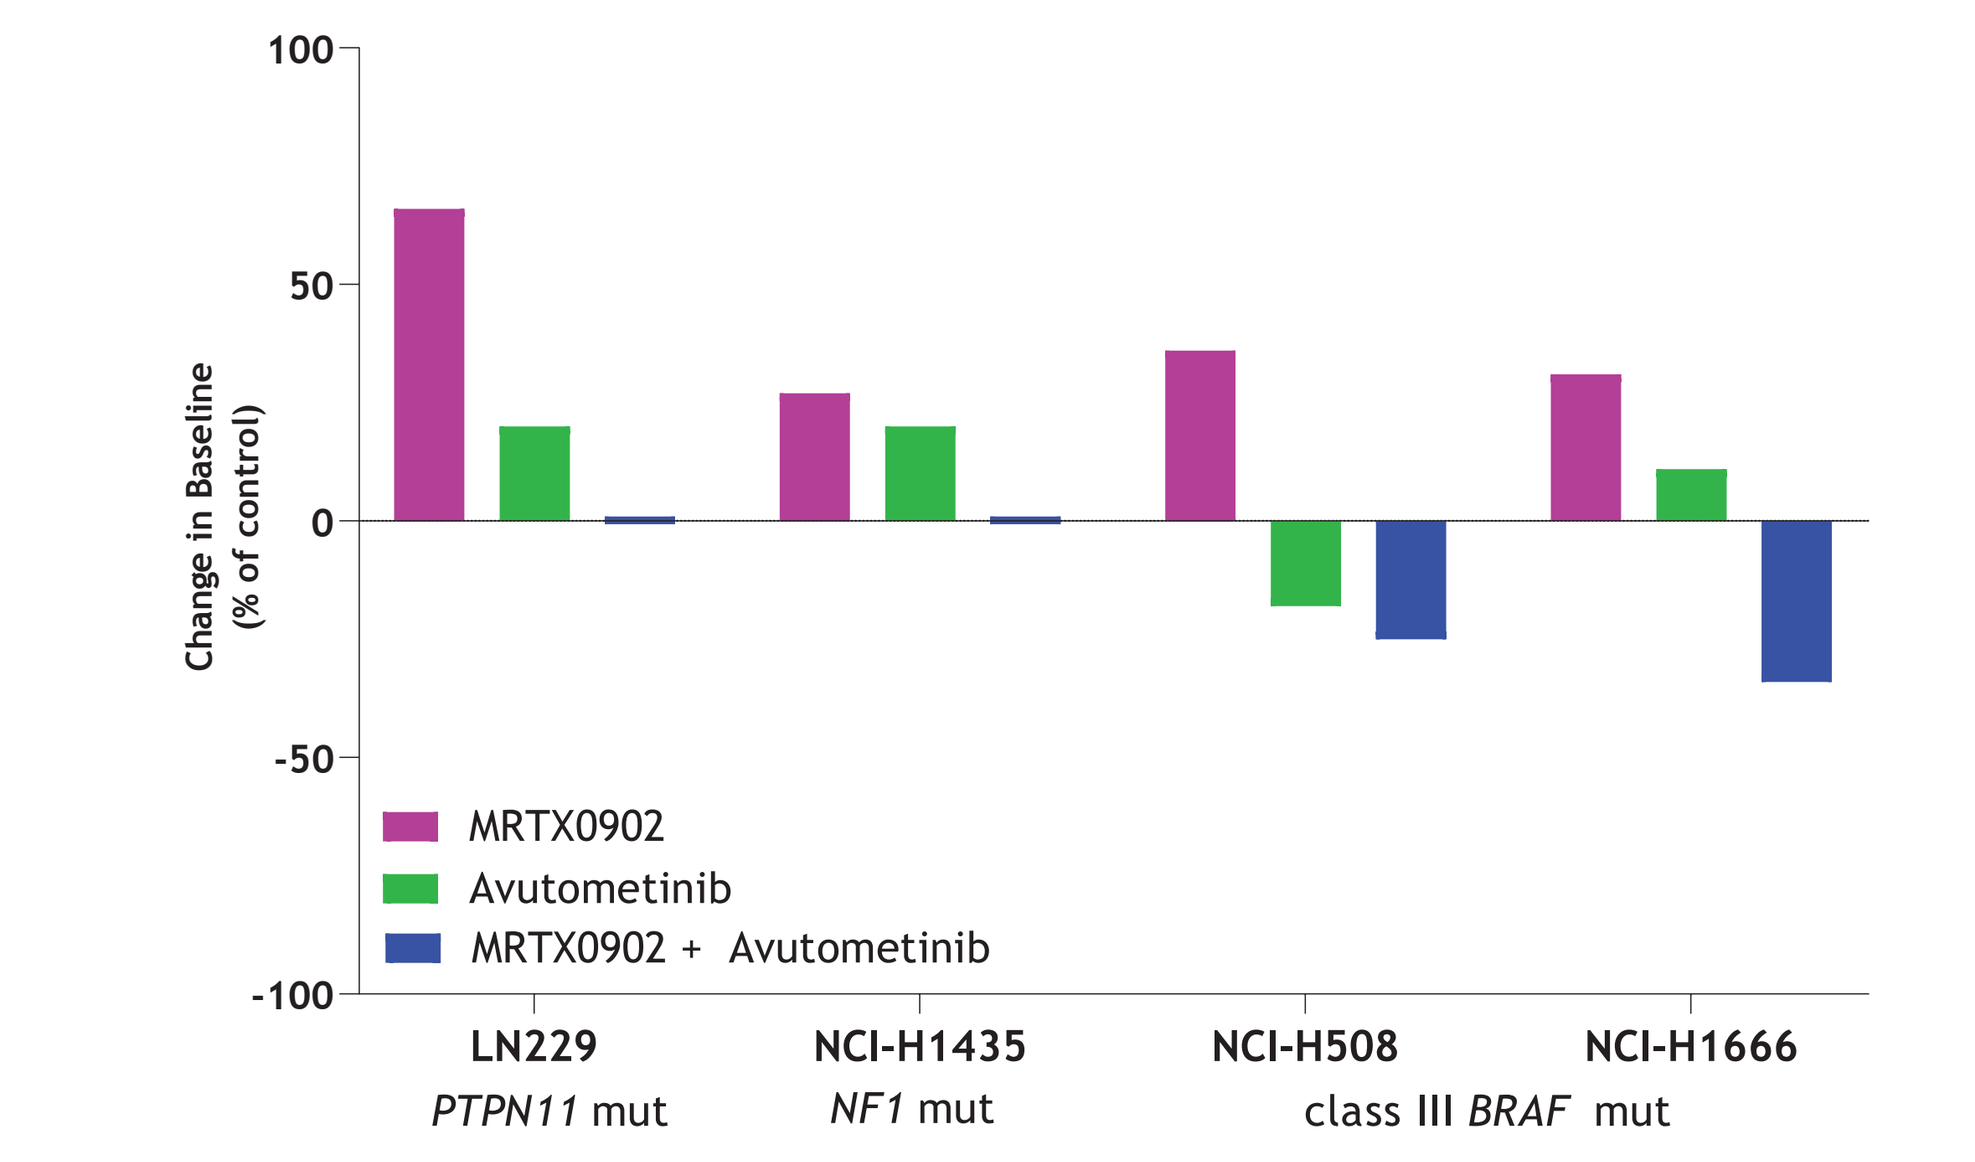

Change in Baseline (% of control)
MRTX0902
Avutometinib
MRTX0902 + Avutometinib
LN229
PTPN11 mut
NCI-H1435
NF1 mut
NCI-H508
NCI-H1666
class III BRAF mut

MRTX0902 was administered twice daily (BID) via oral gavage for ~30-68 days to immunocompromised mice bearing human RAS-MAPK pathway-mutant tumor xenografts (average starting tumor volume of 100-200 mm3) at 50 mg/kg. MRTX0902 was formulated in a suspension of 0.5% methylcellulose (4000 cps) + 0.2% Tween 80 in water. Avutometinib was administered twice daily every 2 days (BID Q2D) via oral gavage for ~30-68 days at 0.3 mg/kg and was formulated in a solution of 5% DMSO:95% HPCD in water. Data for n=5 animals/group is shown as percent change in baseline tumor volume.

Co-administration of MRTX0902 with avutometinib demonstrated increased efficacy and was well-tolerated with no adverse side effects in mice.

CONCLUSIONS

- MRTX0902 is a selective inhibitor of SOS1 currently under evaluation in Phase 1 clinical trials for the treatment of advanced, unresectable, or metastatic cancers with mutations in the KRAS-MAPK pathway (ClinicalTrials.gov Identifier: NCT05578092).
- MRTX0902 potently inhibited RAS-MAPK pathway signaling and proliferation of cancer cell lines harboring mutations in *SOS1*, *NF1*, *PTPN11*, and class III *BRAF*, which translated to robust antitumor activity *in vivo*.
- MRTX0902 augmented the antitumor activity of avutometinib (RAF/MEK clamp) when dosed in combination in select human tumor cell-derived xenograft models harboring mutations in *NF1*, *PTPN11*, and class III *BRAF*. Thus, inhibition of SOS1 is able to address RTK-mediated feedback associated with inhibition of the RAF/MEK node in the RAS-MAPK pathway.
- The mechanism of action and preclinical data support the development of the MRTX0902 and avutometinib combination treatment in patients with RAS-MAPK pathway-dependent cancers characterized by oncogenic mutations in *SOS1*, *PTPN11*, class III BRAF, and *NF1* for which there are currently no approved targeted therapies. Moreover, given the high incidence of RAS-MAPK pathway germline mutations in RASopathies, there is additional therapeutic potential for MRTX0902 in SOS1-dependent genetic syndromes and related cancers.

REFERENCE

1. Prior, IA, et al. *Cancer Res* 2020;80(14):2969-74 doi 10.1158/0008-5472.CAN-19-3682.
2. Corbalan-Garcia, S, et al. *Mol Cell Biol*, 1996. 16(10): p. 5674-82.
3. Simanshu, DK, et al *Biochim Biophys Acta Rev Cancer* 2020;1874(2):188445 doi 10.1016/j.bbcan.2020.188445.
4. Jeng, HH, et al. *Nat Commun* 2012;3:1168 doi 10.1038/ncomms2173.
5. Hillig, RC, et al. *Proc Natl Acad Sci USA* 2019;116(7):2551-60 doi 10.1073/pnas.1812963116.
6. Hofmann, MH, et al. *Cancer Discov* 2021;11(1):142-57 doi 10.1158/2159-8290.CD-20-0142.
7. Ketcham, JM, et al. *J Med Chem* 2022;65(14):9678-90 doi 10.1021/acs.jmedchem.2c00741.
8. Sudhakar, N, et al. *Molecular Cancer Therapeutics* (accepted for publication, February 2024).
9. Ratner, N, et al. *Nat Rev Cancer* 2015;15(5):290-301 doi 10.1038/nrc3911.
10. Consortium APG. *Cancer Discov* 2017;7(8):818-31 doi 10.1158/2159-8290.CD-17-0151.
11. Hart, TC, et al. *Am J Hum Genet* 2002;70(4):943-54 doi 10.1086/339689.
12. Roberts, AE, et al. *Nat Genet* 2007; 39(1):70-4 doi 10.1038/ng1926.
13. Tartaglia, M, et al. *Nat Genet* 2007; 39(1):75-9 doi 10.1038/ng1939.
14. Nichols, RJ, et al. *Nat Cell Biol* 2018; 20(9): 1064-73 doi 10.1038/s41556-018-0169-1.
15. Liu, Y, et al. *Cellular and Molecular Life Sciences* 2023;doi 10.1007/s00018-023-05052-8.
16. Hallin, J, et al., *Cancer Discov* 2020; 10(1):54-71 doi 10.1158/2159-8290.CD-19-1167.

ACKNOWLEDGMENTS

- This study was sponsored by Mirati, a Bristol Myers Squibb company.

Scientific Content on Demand

To request a copy of this poster

Scan QR code via a barcode reader application

QR codes are valid for 30 days after the congress presentation date

Presented at the American Association for Cancer Research (AACR) Annual Meeting; April 5-10, 2024; San Diego, CA

Email: sudhakarn@mirati.com

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.