Phase 3 Trial of Sitravatinib plus Nivolumab vs Docetaxel for Treatment of NSCLC After Platinum-Based Chemotherapy and Immunotherapy (SAPPHIRE)

Ivor John Percent[1], Craig H. Reynolds[2], Kartik Konduri[3], Matthew Thomas Whitehurst[4], Emmanuel A. Nidhiry[5], Ronald H. Yanagihara[6], Misako Nagasaka[7], Marshall T Schreeder[8], James Uyeki[9], Georges Azzi[10], Krishna Kishore Pachipala[11], Jason C. Comer[12], Rachel E. Lerner[13], Ticiana Leal[14], Delia Alvarez[15], Saskia T.C. Neuteboom[15], Natasha Reddinger[15], Xiaohong Yan[15], Ronald L. Shazer[15], David Michael Waterhouse[16]

[1]Florida Cancer Specialists South/Sarah Cannon Research Institute, Port Charlotte, FL; [2]Florida Cancer Affiliates, Ocala, FL; [3]Texas Oncology/The US Oncology Network, Dallas, TX; [4]Hematology Oncology Associates of Fredericksburg, Fredericksburg, VA; [5]Commonwealth Cancer Ctr, Danville, KY; [6]Straub Clinic, Honolulu, HI; [7]Barbara Ann Karmanos Cancer Institute, Detroit, MI; [8]Clearview Cancer Institute, Huntsville, AL; [9]Texas Oncology PA, Austin, TX; [10]Holy Cross Medical Group, Michael and Dianne Bienes Comprehensive Cancer Center, Fort Lauderdale, FL; [11]Millenium Physicians-North Houston, Houston, TX; [12]Overlake Medical Clinics, Bellevue, WA; [13]Frauenshuh Cancer Center and Park Nicollet Institute, Minneapolis, MN; [14]University of Wisconsin Carbone Cancer Center, Madison, WI; [15]Mirati Therapeutics, Inc., San Diego, CA; Mirati, San Diego, CA; [16]OHC (Oncology Hematology Care), Inc., Cincinnati, OH

BACKGROUND

Checkpoint Inhibitor Therapy (CIT) for Advanced Lung Cancer

- Non-small cell lung cancer (NSCLC) accounts for approximately 83% of all lung cancer cases, of which approximately half are classified as adenocarcinoma of the lung
- CIT has improved Overall Survival (OS) outcomes relative to historic standards of care across many tumor types
- However, some patients initially derive benefit from anti PD-1/L1 based therapy, but eventually relapse
- Effective treatment options are needed to improve long-term survival in this patient population

Sitravatinib in the Tumor Microenvironment (TME)

- Sitravatinib is an orally available, small molecule inhibitor of several closely related Receptor Tyrosine Kinases (RTKs) including the TAM (TYRO3, AXL and MERTK) and split (VEGFR2, KIT) family receptors, which regulate the immunosuppressive cell types implicated in resistance to checkpoint inhibition

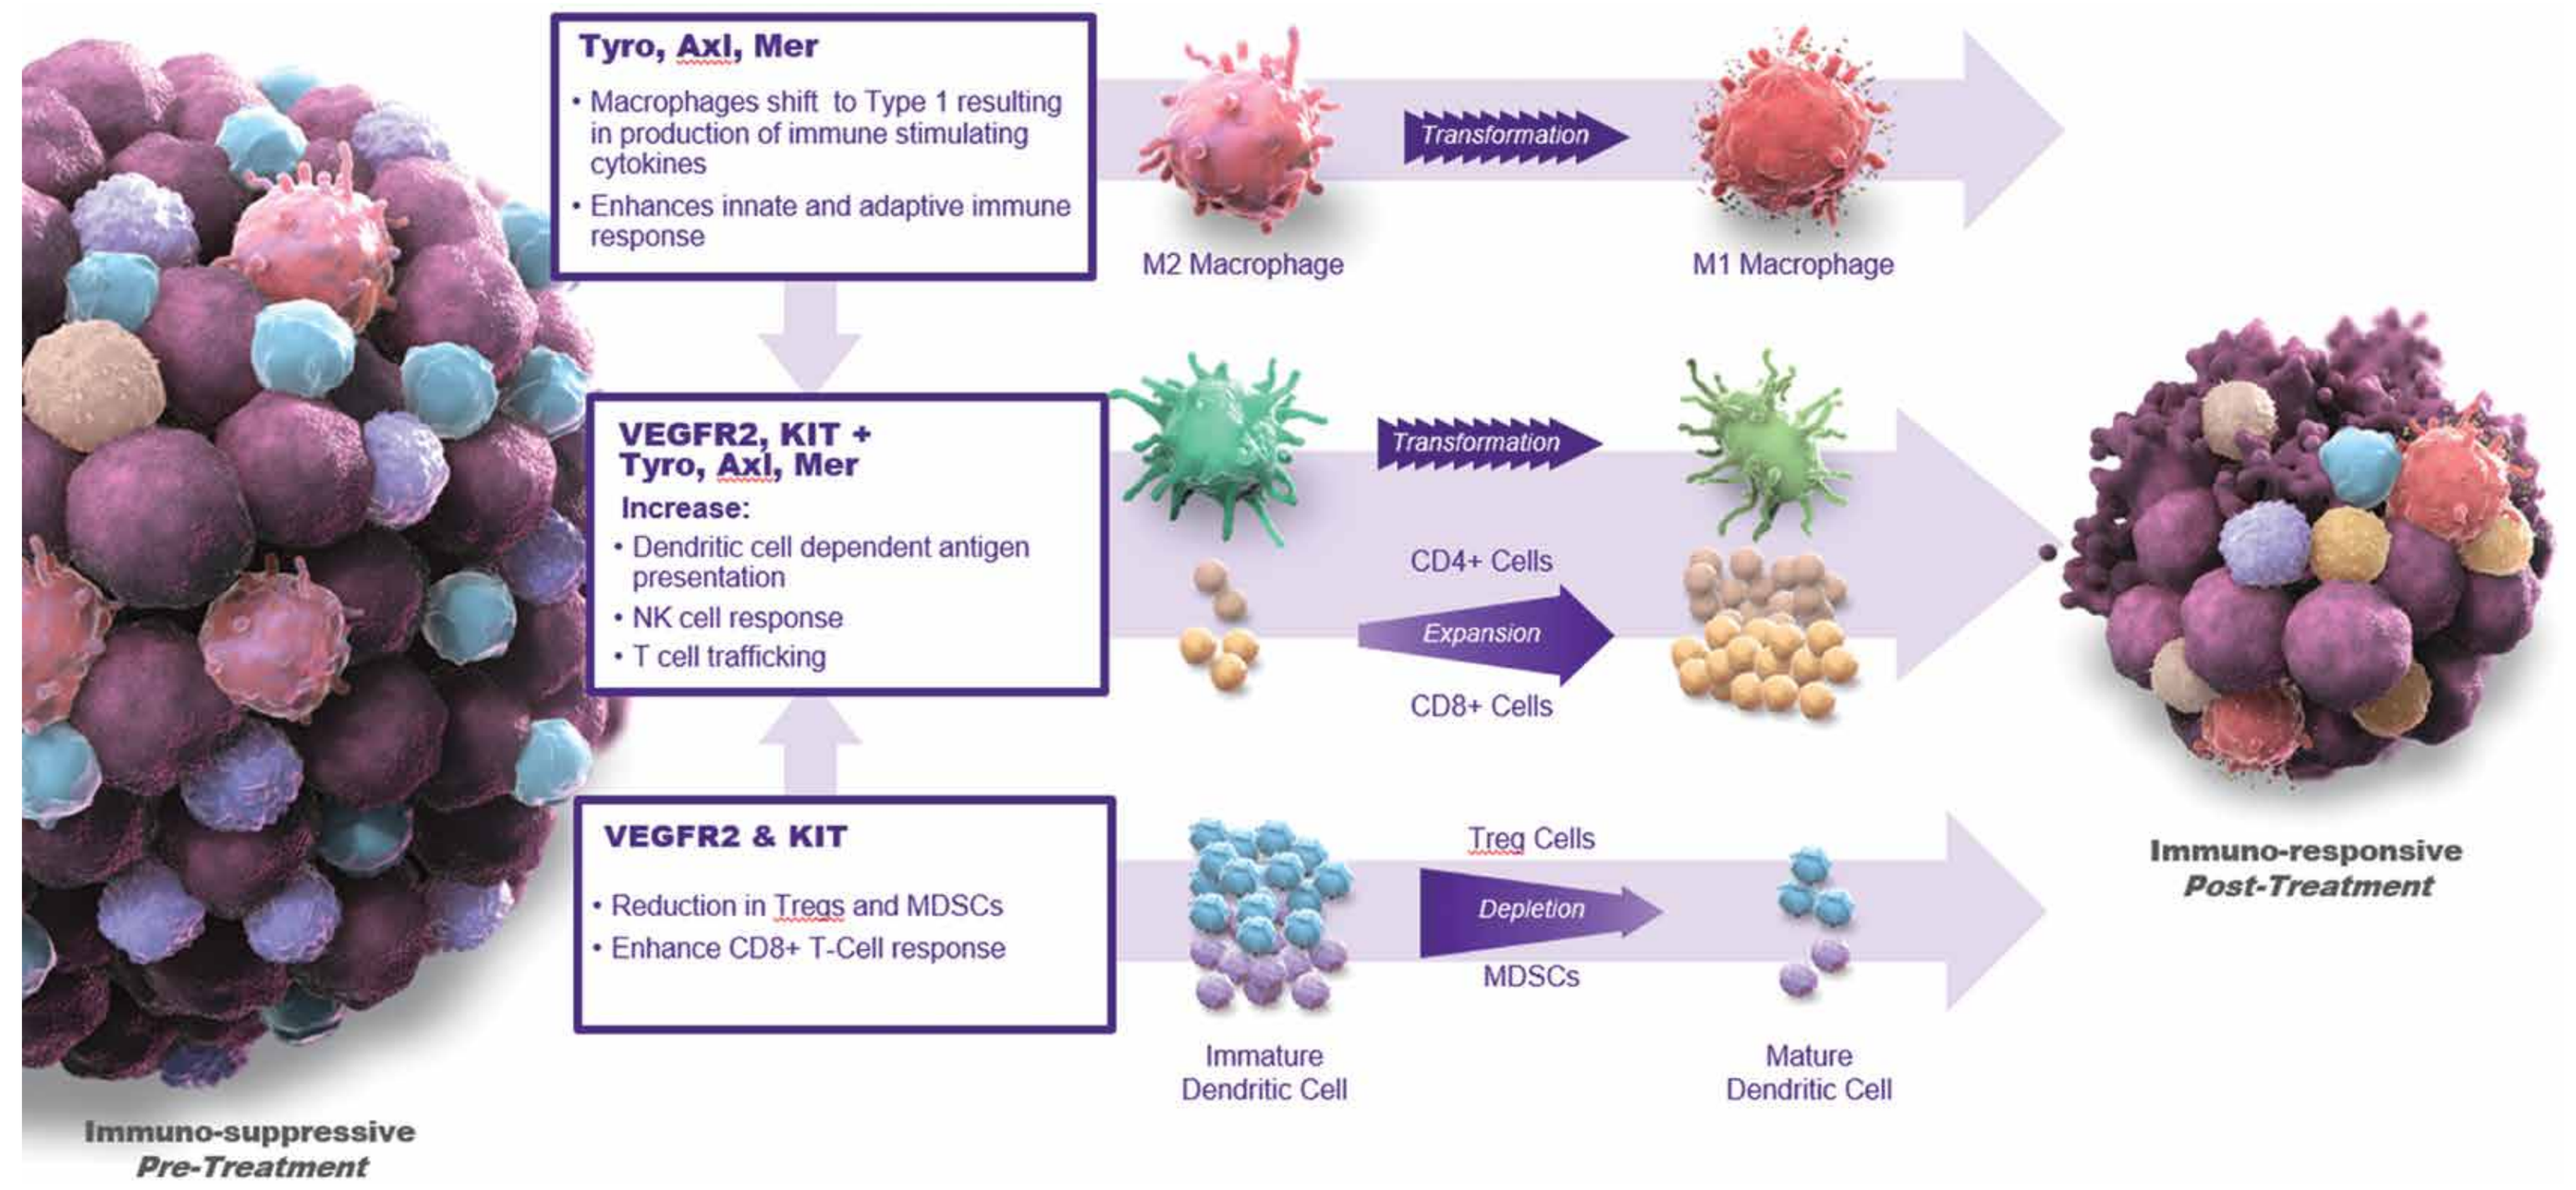
Tyro, Axl, Mer
• Macrophages shift to Type 1 resulting in production of immune stimulating cytokines
• Enhances innate and adaptive immune response
M2 Macrophage
Transformation
M1 Macrophage
VEGFR2, KIT + Tyro, Axl, Mer
Increase:
• Dendritic cell dependent antigen presentation
• NK cell response
• T cell trafficking
Transformation
CD4+ Cells
Expansion
CD8+ Cells
VEGFR2 & KIT
• Reduction in Tregs and MDSCs
• Enhance CD8+ T-Cell response
Treg Cells
Depletion
MDSCs
Immature Dendritic Cell
Mature Dendritic Cell
Immuno-suppressive Pre-Treatment
Immuno-responsive Post-Treatment

Figure 1. Sitravatinib in the Tumor Microenvironment (TME)
There is evidence that sitravatinib sensitizes tumors to immune CIT

Sitravatinib Sensitizes the Tumor Microenvironment to Anti-Tumor Immune Response

- Tumor tolerance/resistance to checkpoint inhibition may be mediated by multiple immunosuppressive immune cell types in the tumor microenvironment:
 1. Regulatory T cells (Tregs)
 2. Myeloid-derived suppressor cells (MDSCs)
 3. M2-polarized macrophages
- Sitravatinib may reverse resistance to PD-1/L1 inhibitors by decreasing immuno-suppressive cells in the TME
- Preliminary data from a Phase 1 study of sitravatinib and nivolumab in patients with oral cavity cancer (SNOW study), demonstrated that sitravatinib led to a decrease in MDSCs and a shift towards M1-type macrophages in the TME in some patients with oral cavity squamous cell carcinoma; most patients had clinical benefit and tumor downstaging, with none requiring post operative chemotherapy
- Combination of sitravatinib with PD-1/L1 inhibitor is a rational approach to augmenting the anti-tumor immune response and improving outcomes by overcoming resistance to anti PD-1/L1 therapy

BACKGROUND

Encouraging Preliminary OS Data from Phase 2 Trial MRTX-500: Sitravatinib + Nivolumab in NSCLC patients who progressed on or after CIT

- Preliminary median OS of 18.1 months for the subset of Prior Clinical Benefit (PCB) patients who received the combination as either 2nd or 3rd line of therapy after progressing on treatment with a CIT (n=73) **(Figure 2)**
- This patient cohort is consistent with the inclusion criteria for the ongoing Phase 3 SAPPHIRE study
- Historical OS for Docetaxel in 2nd Line: 8.5 – 9.6 months

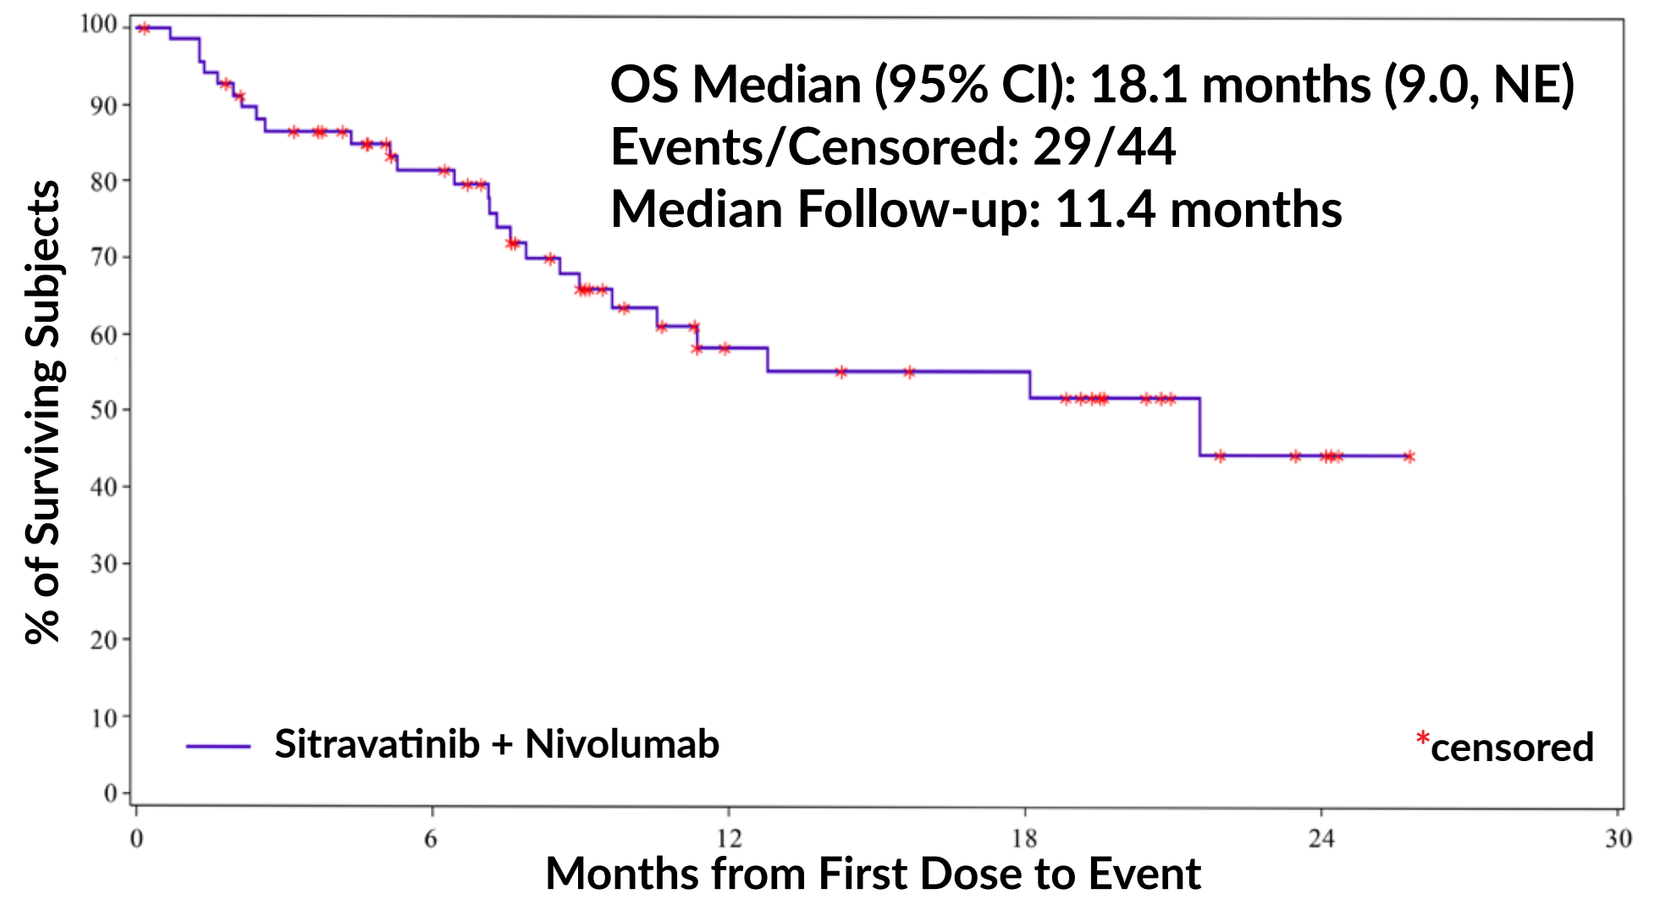
OS Median (95% CI): 18.1 months (9.0, NE)
Events/Censored: 29/44
Median Follow-up: 11.4 months
% of Surviving Subjects
100
90
80
70
60
50
40
30
20
10
0
Sitravatinib + Nivolumab
*censored
0
6
12
18
24
30
Months from First Dose to Event

Figure 2. MRTX-500: Overall Survival, PCB Cohort: 1 or 2 Prior Lines of Treatment, N=73, Data as of January 30, 2020

STUDY OBJECTIVES

PRIMARY OBJECTIVE

To compare OS in patients with non-squamous NSCLC, who have experienced disease progression on or after platinum-based chemotherapy and checkpoint inhibitor therapy, treated with sitravatinib and nivolumab versus docetaxel

SECONDARY OBJECTIVES

- To evaluate safety and tolerability of sitravatinib in combination with nivolumab
- To evaluate secondary efficacy endpoints including ORR per RECIST 1.1, DOR, CBR, PFS, 1-Year Survival Rate
- To evaluate the pharmacokinetics (PK) of sitravatinib administered in combination with nivolumab
- To evaluate health-related quality of life and lung cancer-specific symptoms

EXPLORATORY OBJECTIVES

- To assess correlations between baseline tumor immune biomarkers and gene mutations and treatment-related outcomes
- To evaluate efficacy endpoints using iRECIST
- To characterize the immunogenicity of nivolumab in combination with sitravatinib

METHODS

STUDY DESIGN

- Global, randomized (1:1), open-label, Phase 3 study of sitravatinib in combination with nivolumab vs docetaxel in patients with advanced non-squamous NSCLC who have progressed on or after CIT and platinum based chemotherapy. **(Figure 3)**

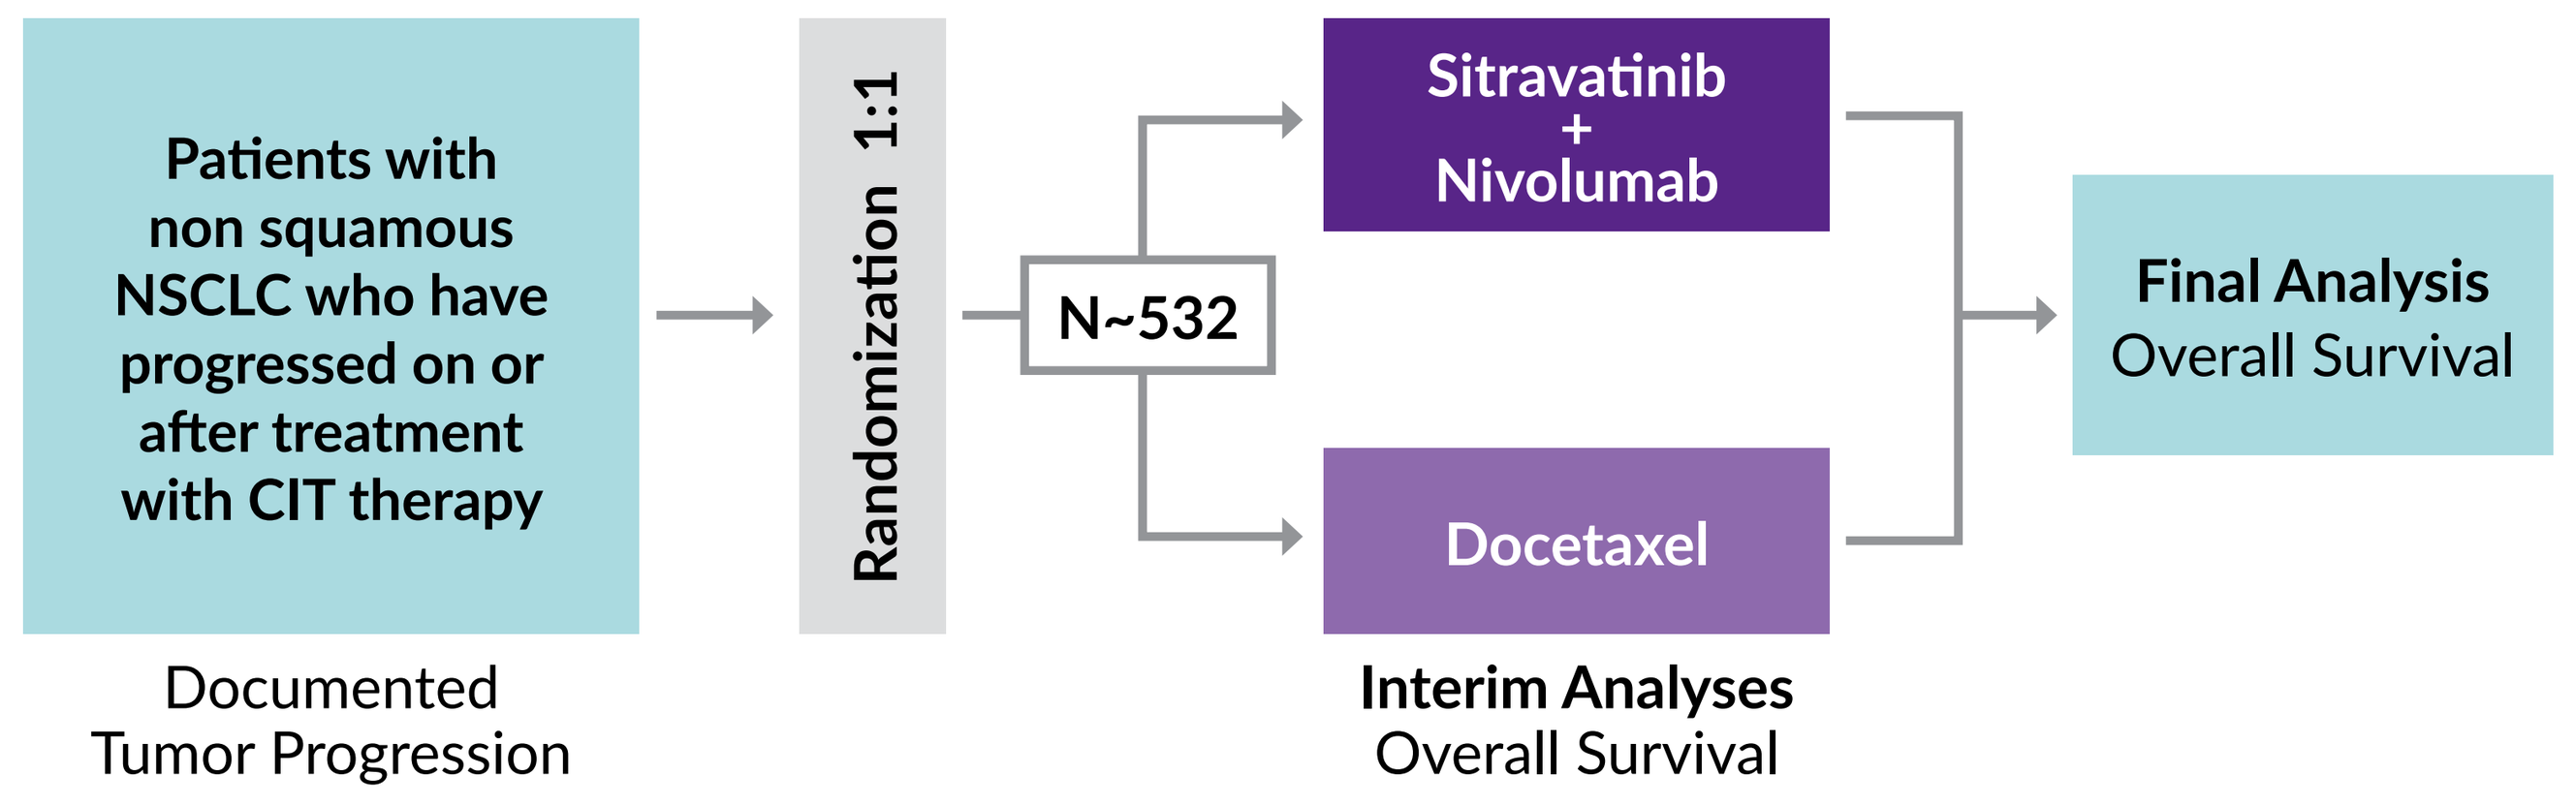
Patients with non squamous NSCLC who have progressed on or after treatment with CIT therapy
Documented Tumor Progression
Randomization 1:1
N~532
Sitravatinib + Nivolumab
Docetaxel
Interim Analyses
Overall Survival
Final Analysis
Overall Survival

Stratification Factors:
- Prior treatment regimens in the advanced setting (1 vs 2)
- ECOG performance status (0 vs 1)
- Brain metastasis (presence vs absence)

Figure 3. SAPPHIRE Study Design

METHODS

KEY INCLUSION CRITERIA

- Histologically or cytologically confirmed **non-squamous NSCLC** with metastatic (Stage IV) or unresectable, locally advanced (Stage IIIB/IIIC) disease, not amenable to treatment with curative intent
- Receipt of **at least one but not more than two** prior treatment regimens in the advanced disease setting to include:
 - Treatment with a CIT (i.e., anti-PD-1/L1) and a platinum-based chemotherapy*, which may have been in combination or in sequence (i.e., platinum-based chemotherapy followed by CIT)
 - Prior treatment may have included maintenance therapy with a chemotherapy agent (e.g., pemetrexed) and/or a CIT
 - Most recent treatment regimen must have included a CIT with radiographic disease progression on or after treatment
- Duration of treatment on most recent CIT of at least 4 months without disease progression during this 4-month period
- Adequate bone marrow and organ function

** Platinum-based adjuvant, neoadjuvant, or definitive chemoradiation therapy given for locally advanced disease followed by recurrent or metastatic disease within 6 months of completing therapy is considered treatment in the advanced disease setting*

KEY EXCLUSION CRITERIA

- Discontinuation of prior treatment with CIT more than 90 days prior to the date of randomization
- Receipt of systemic cancer therapy since discontinuation of CIT, with the exception of maintenance chemotherapy
- Active brain metastases, unless asymptomatic
- Prior therapies:
 - Immunotherapies not previously specified, including anti-CTLA-4, anti-OX40 and anti–CD137
 - Cancer therapy having the same mechanism of action as sitravatinib (e.g., tyrosine kinase inhibitor with a similar target profile or bevacizumab)
- Tumors that test positive for EGFR, ROS1, ALK mutations, or ALK fusions
- Unacceptable toxicity on prior CIT
- Active or prior documented autoimmune disease
- Active or prior immunocompromising conditions
- Impaired heart function

STUDY TREATMENTS

- Sitravatinib + Nivolumab arm
 - Cycle length 28 days
 - Oral sitravatinib once daily in combination with intravenous nivolumab 240mg every 2 weeks or 480mg every 4 weeks
- Docetaxel arm
 - Cycle length 21 days
 - Intravenous docetaxel 75mg/m^2 every 3 weeks

ASSESSMENTS

- Routine safety assessments performed throughout the study
- Disease assessments per RECIST version 1.1
- PK parameters evaluated after administration of sitravatinib in combination with nivolumab
- Other assessments include patient reported outcomes, baseline tumor PD-L1 expression, gene alterations in tumor tissue (optional) and in circulating tumor DNA (optional)

STUDY SITES

- 211 sites in 12 countries **(Figure 4)**

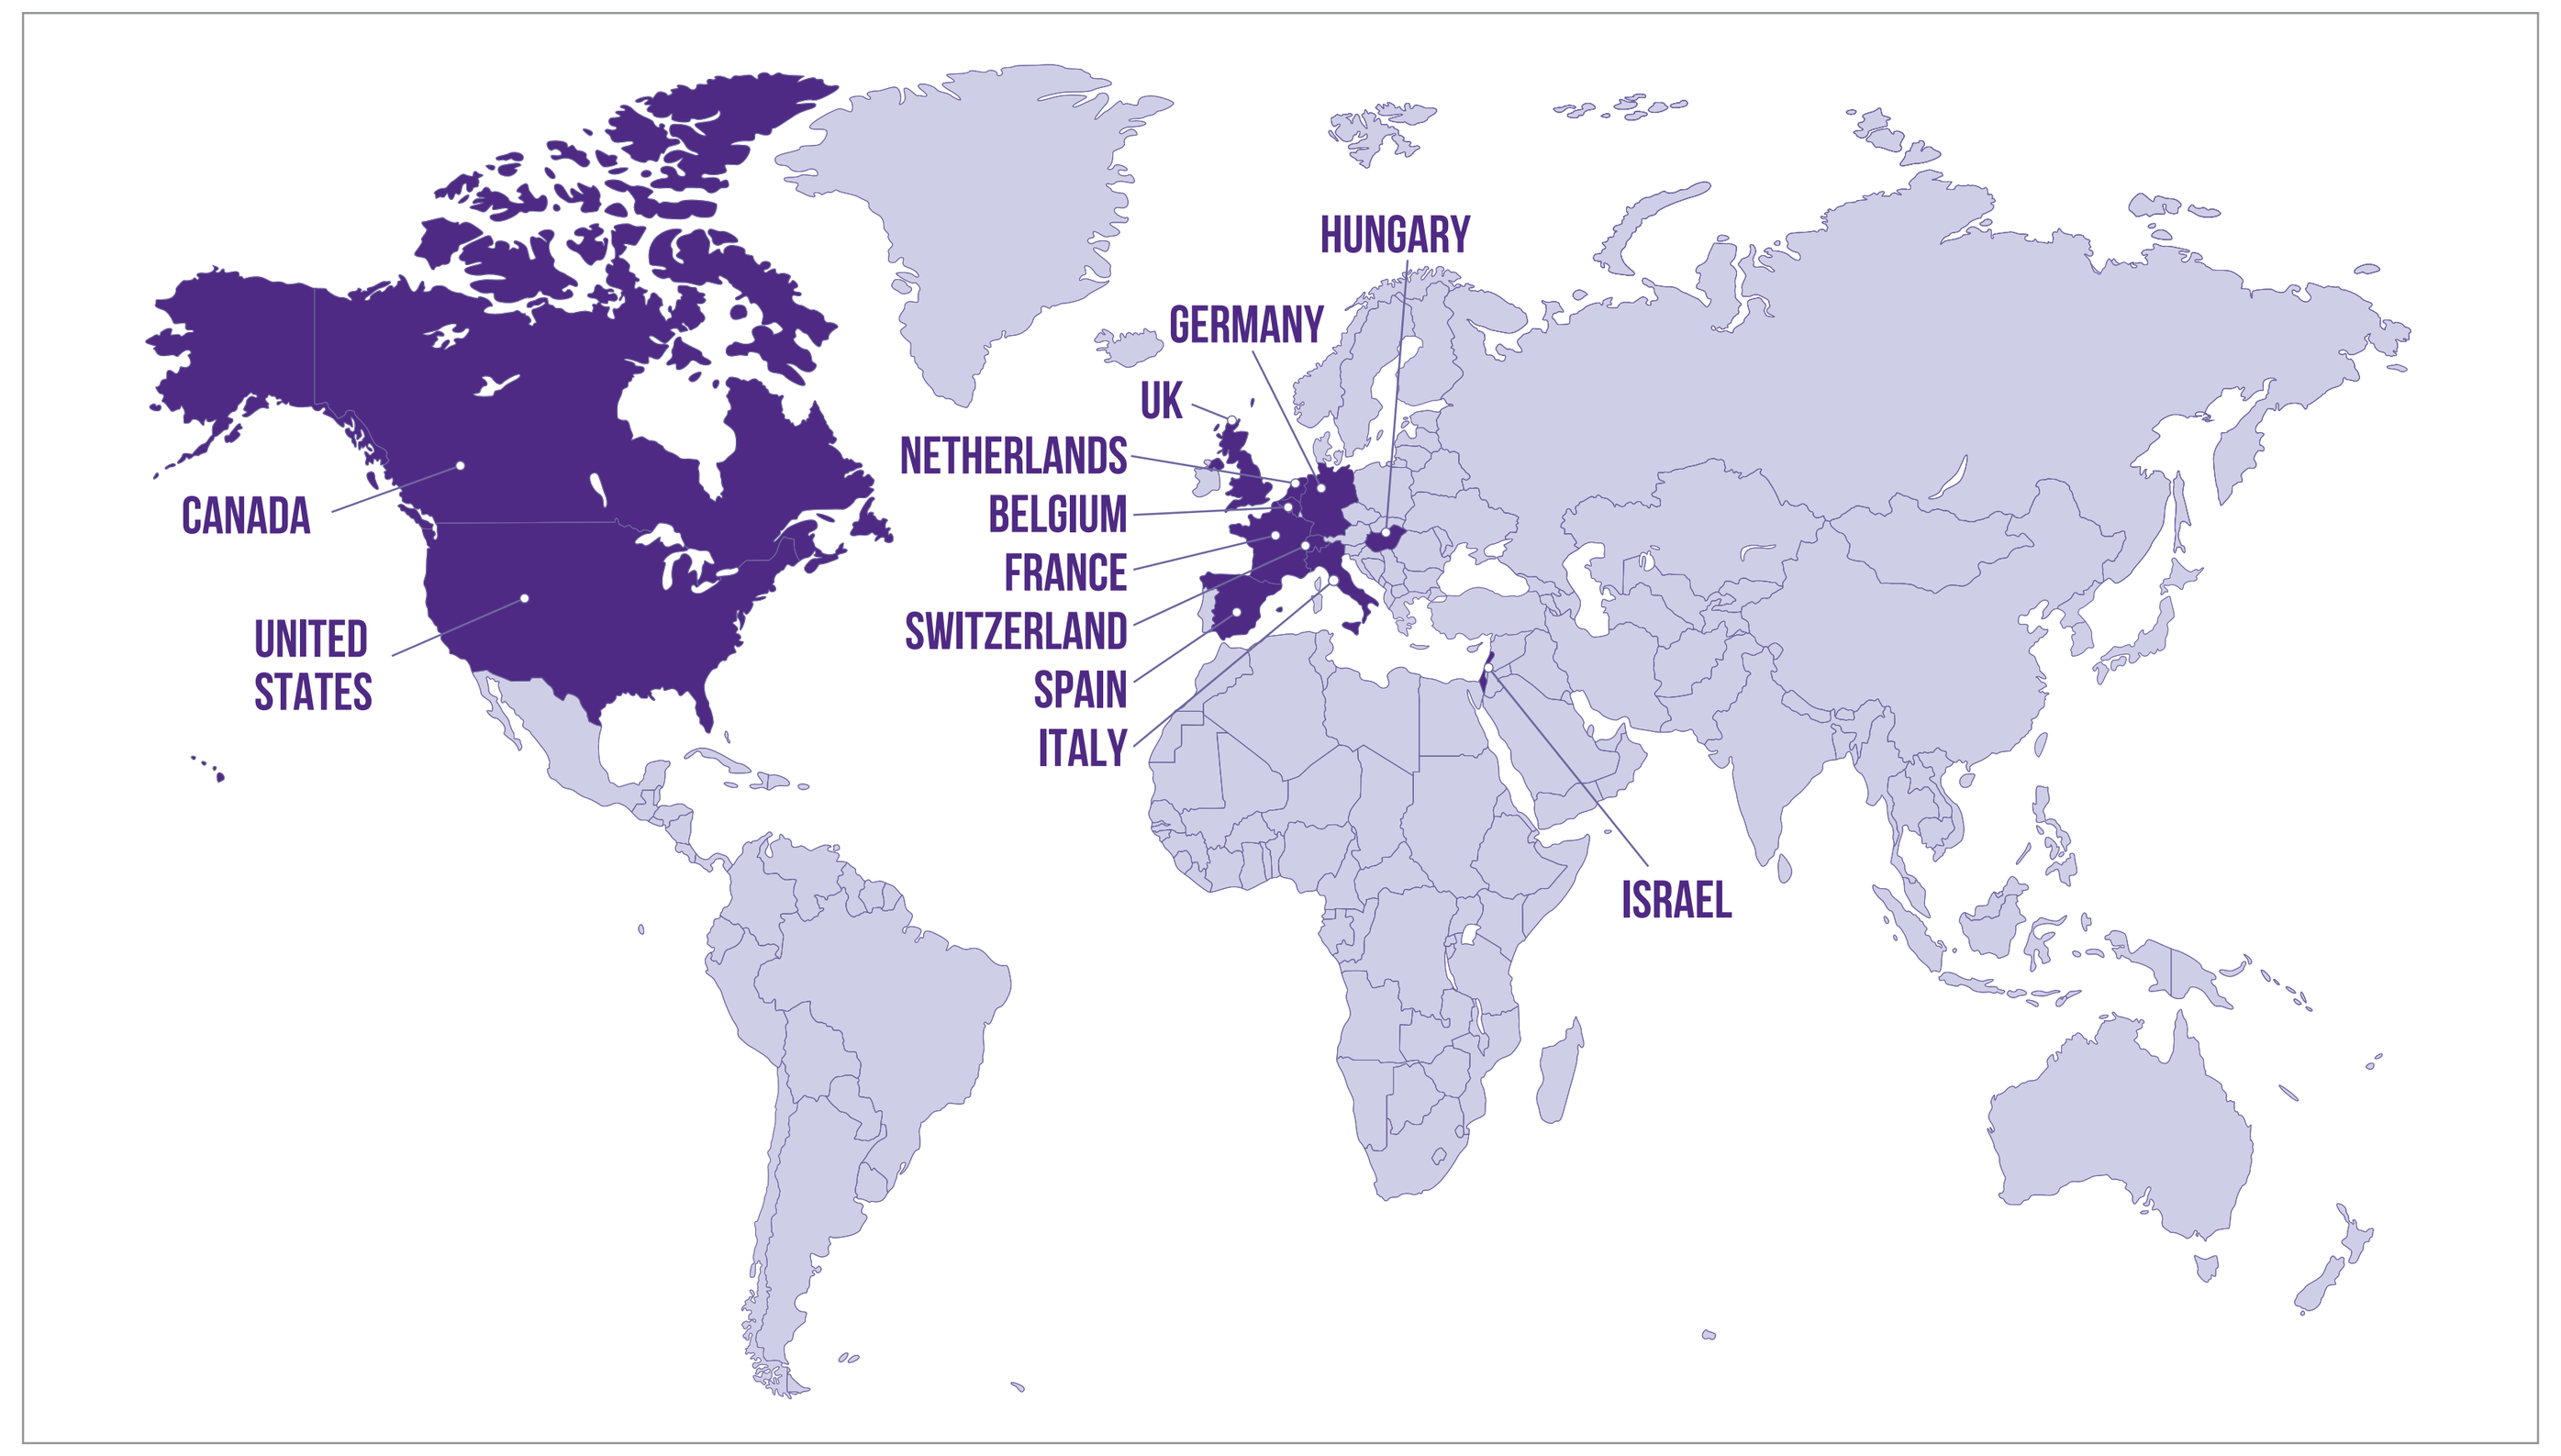
HUNGARY
GERMANY
UK
NETHERLANDS
BELGIUM
FRANCE
SWITZERLAND
SPAIN
ITALY
CANADA
UNITED STATES
ISRAEL

Figure 4. Study Sites

SUMMARY

- Despite advances with anti PD-1/L1 therapy, most patients with advanced NSCLC have incurable disease; additional effective treatment options remain limited, particularly after failure and resistance to PD-1/L1 inhibitors
- Effective treatment options are needed to improve long-term survival in this patient population
- The combination of sitravatinib with nivolumab is a rational approach to restoring or enhancing the clinical activity of CIT in patients with immunotherapy resistant NSCLC
- Early signs of clinical activity from the MRTX-500 study have been observed in patients with disease progression following prior anti PD-1/L1 therapy, and support the rationale for the Phase 3 SAPPHIRE study
- The Phase 3 SAPPHIRE study, evaluating sitravatinib plus nivolumab versus docetaxel in NSCLC after platinum-based chemotherapy and immunotherapy is open for enrollment and recruitment is ongoing
- Clinical trial registry number: NCT03906071

REFERENCES

1. Noone AM, et al. SEER Cancer Statistics Rev, 1975-2015; April 2018
2. Oliva M, et al. 34th annual SITC conference; 2019 Nov 6-10; National Harbor, MD
3. Borghaei H, et al. *N Engl J Med*. 2015;373:1627-1639
4. Rittmeyer A, et al. *Lancet*. 2017;389:255-265
5. Herbst RS, et al. *Lancet*. 2016;387:1540-1550
6. https://ir.mirati.com/news-releases/news-details/2020/Mirati-Therapeutics-Reports-First-Quarter-2020-Financial-Results-And-Recent-Corporate-Updates/default.aspx

ACKNOWLEDGEMENT

- Nivolumab provided through collaboration with Bristol Myers Squibb

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Study Trial Information:
www.mirati.com
Email:
516-005@mirati.com